30 days and one year impact of the antithrombotic treatment following TAVI

Pauline BALAGNY[1], Paul GUEDENEY[1], Guillaume LEBRETON[1], Mojgan LAALI[1], Hélène Rousseau[1], Eric VICAUT[1], Sadia MOHAMMEDI[1], Olivier BARTHELEMY[1], Gilles MONTALESCOT[1], Pascal LEPRINCE[1], Jean-Philippe COLLET[1].

[1]Université Pierre et Marie Curie (UPMC Paris 6), ACTION Study Group (www.action-coeur.org), INSERM UMRS-1166, Institut de Cardiologie, Pitié-Salpêtrière Hospital (AP-HP), France

BACKGROUND AND PURPOSE

- Optimal antithrombotic regimen after TAVI is not evidence-based and its impact on outcome is debated.
- To evaluate clinical outcome after successful TAVI according to anti-thrombotic treatment at discharge.

METHODS

- Single-center study between 2010 and 2014
- 2 groups : patients previously or discharged on oral anticoagulation (OAC) and patients discharged on single or dual antiplatelet therapy (SAPT, DAPT) alone
- Primary endpoint : VARC-2's safety endpoint at 30 days combining all-cause mortality, major stroke, life-threatening bleeding, acute kidney injury, myocardial infraction, major vascular complication and repeat procedure for valve-related dysfunction
- Secondary endpoint : VARC-2's efficacy endpoint at 1 year combining all-cause mortality (after 30 days), hospitalization for symptoms of valve-related or cardiac decompensation and prosthetic heart valve dysfunction

RESULTS

Flow chart

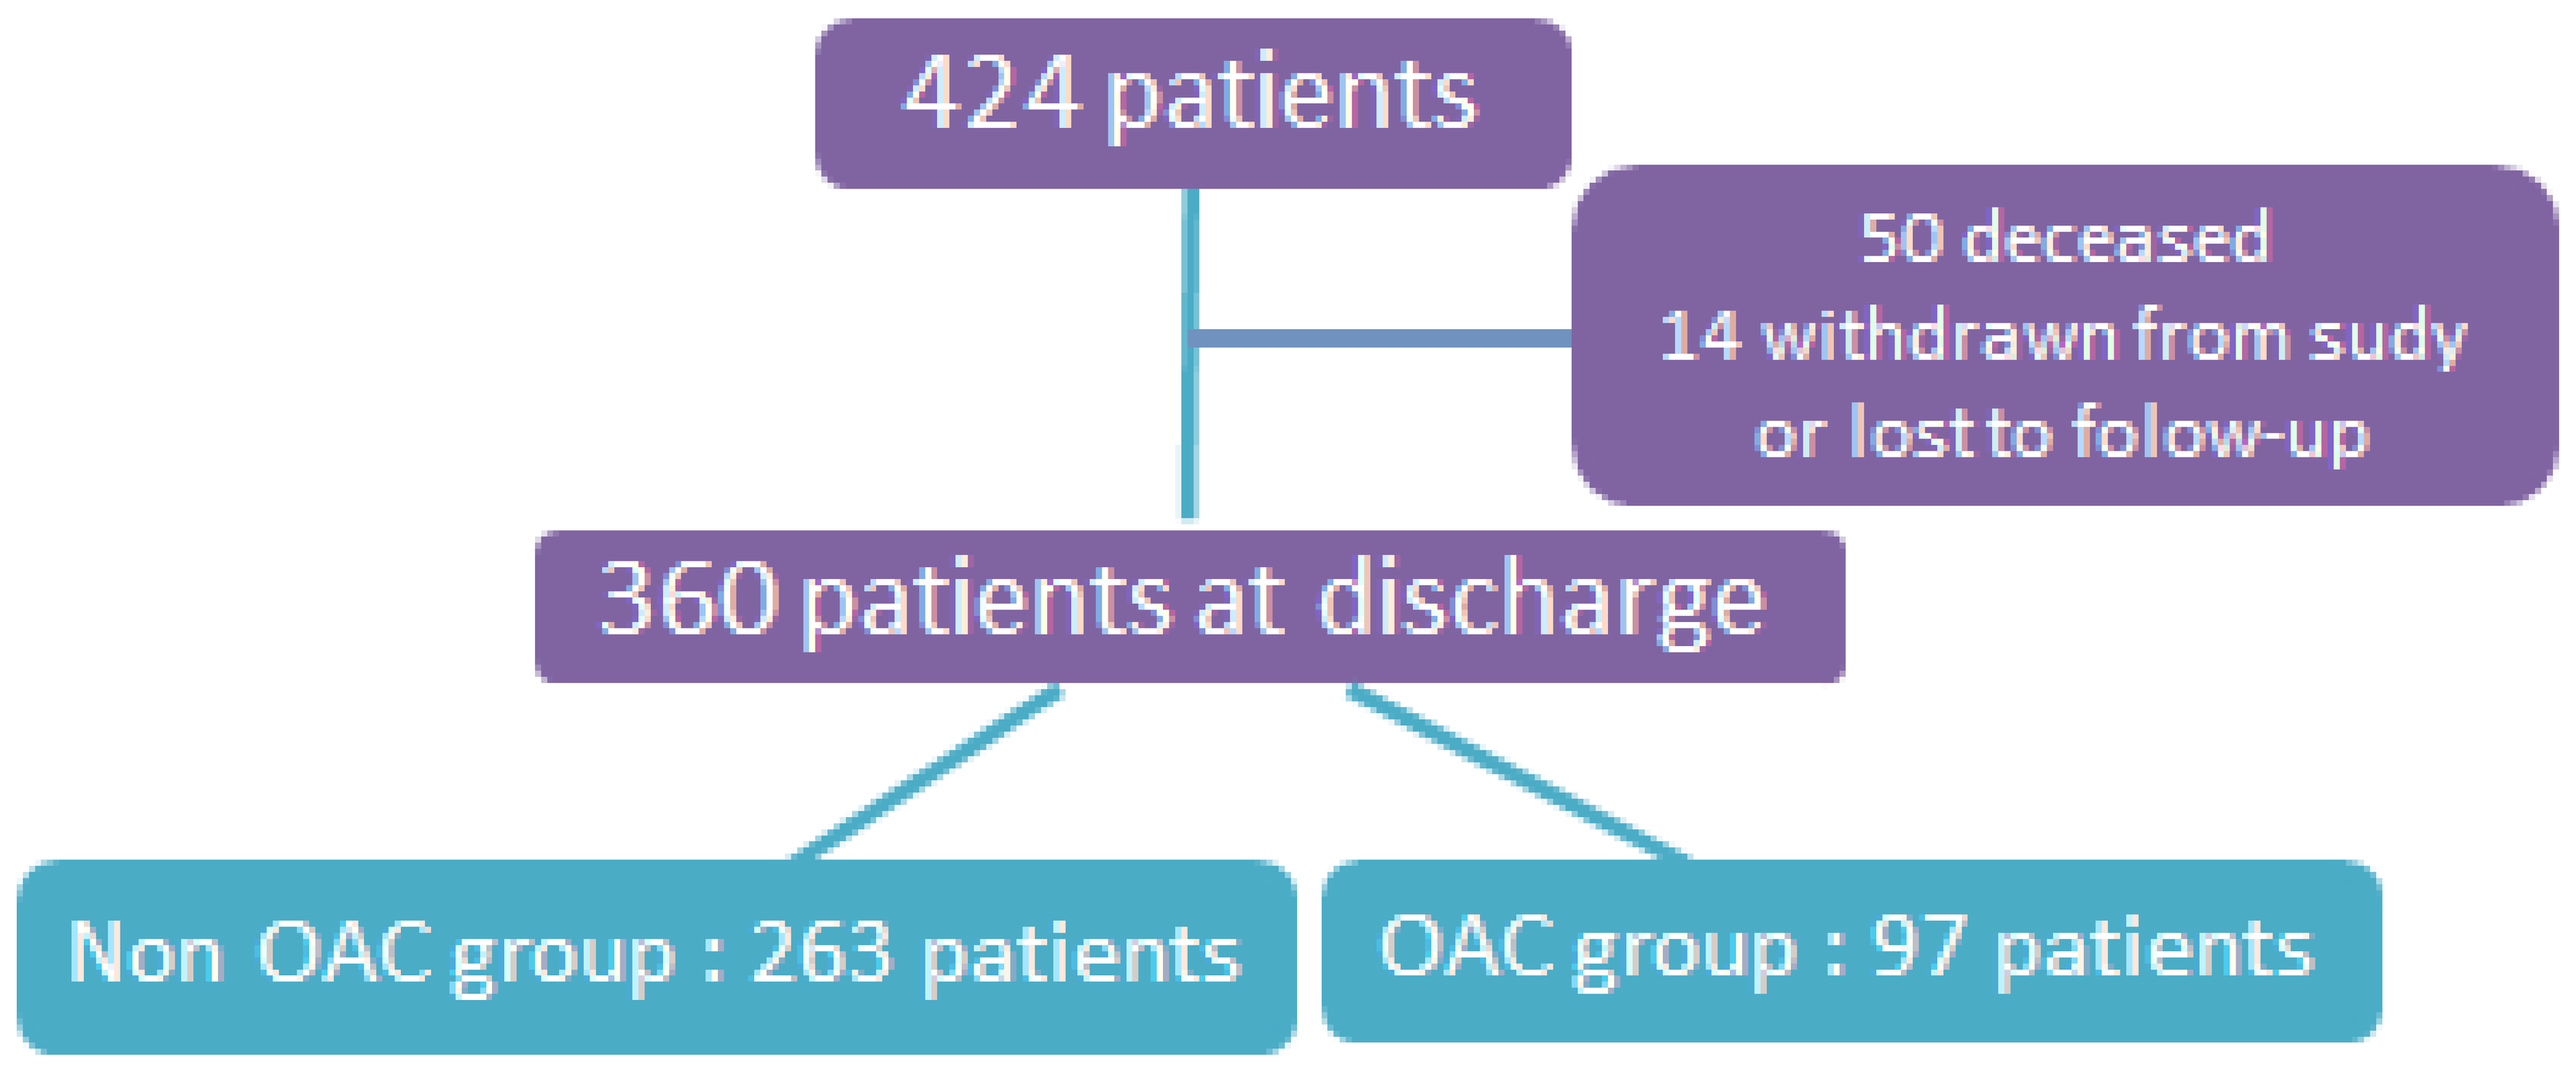
424 patients
50 deceased
14 withdrawn from sudy
or lost to folow-up
360 patients at discharge
Non OAC group : 263 patients
OAC group : 97 patients

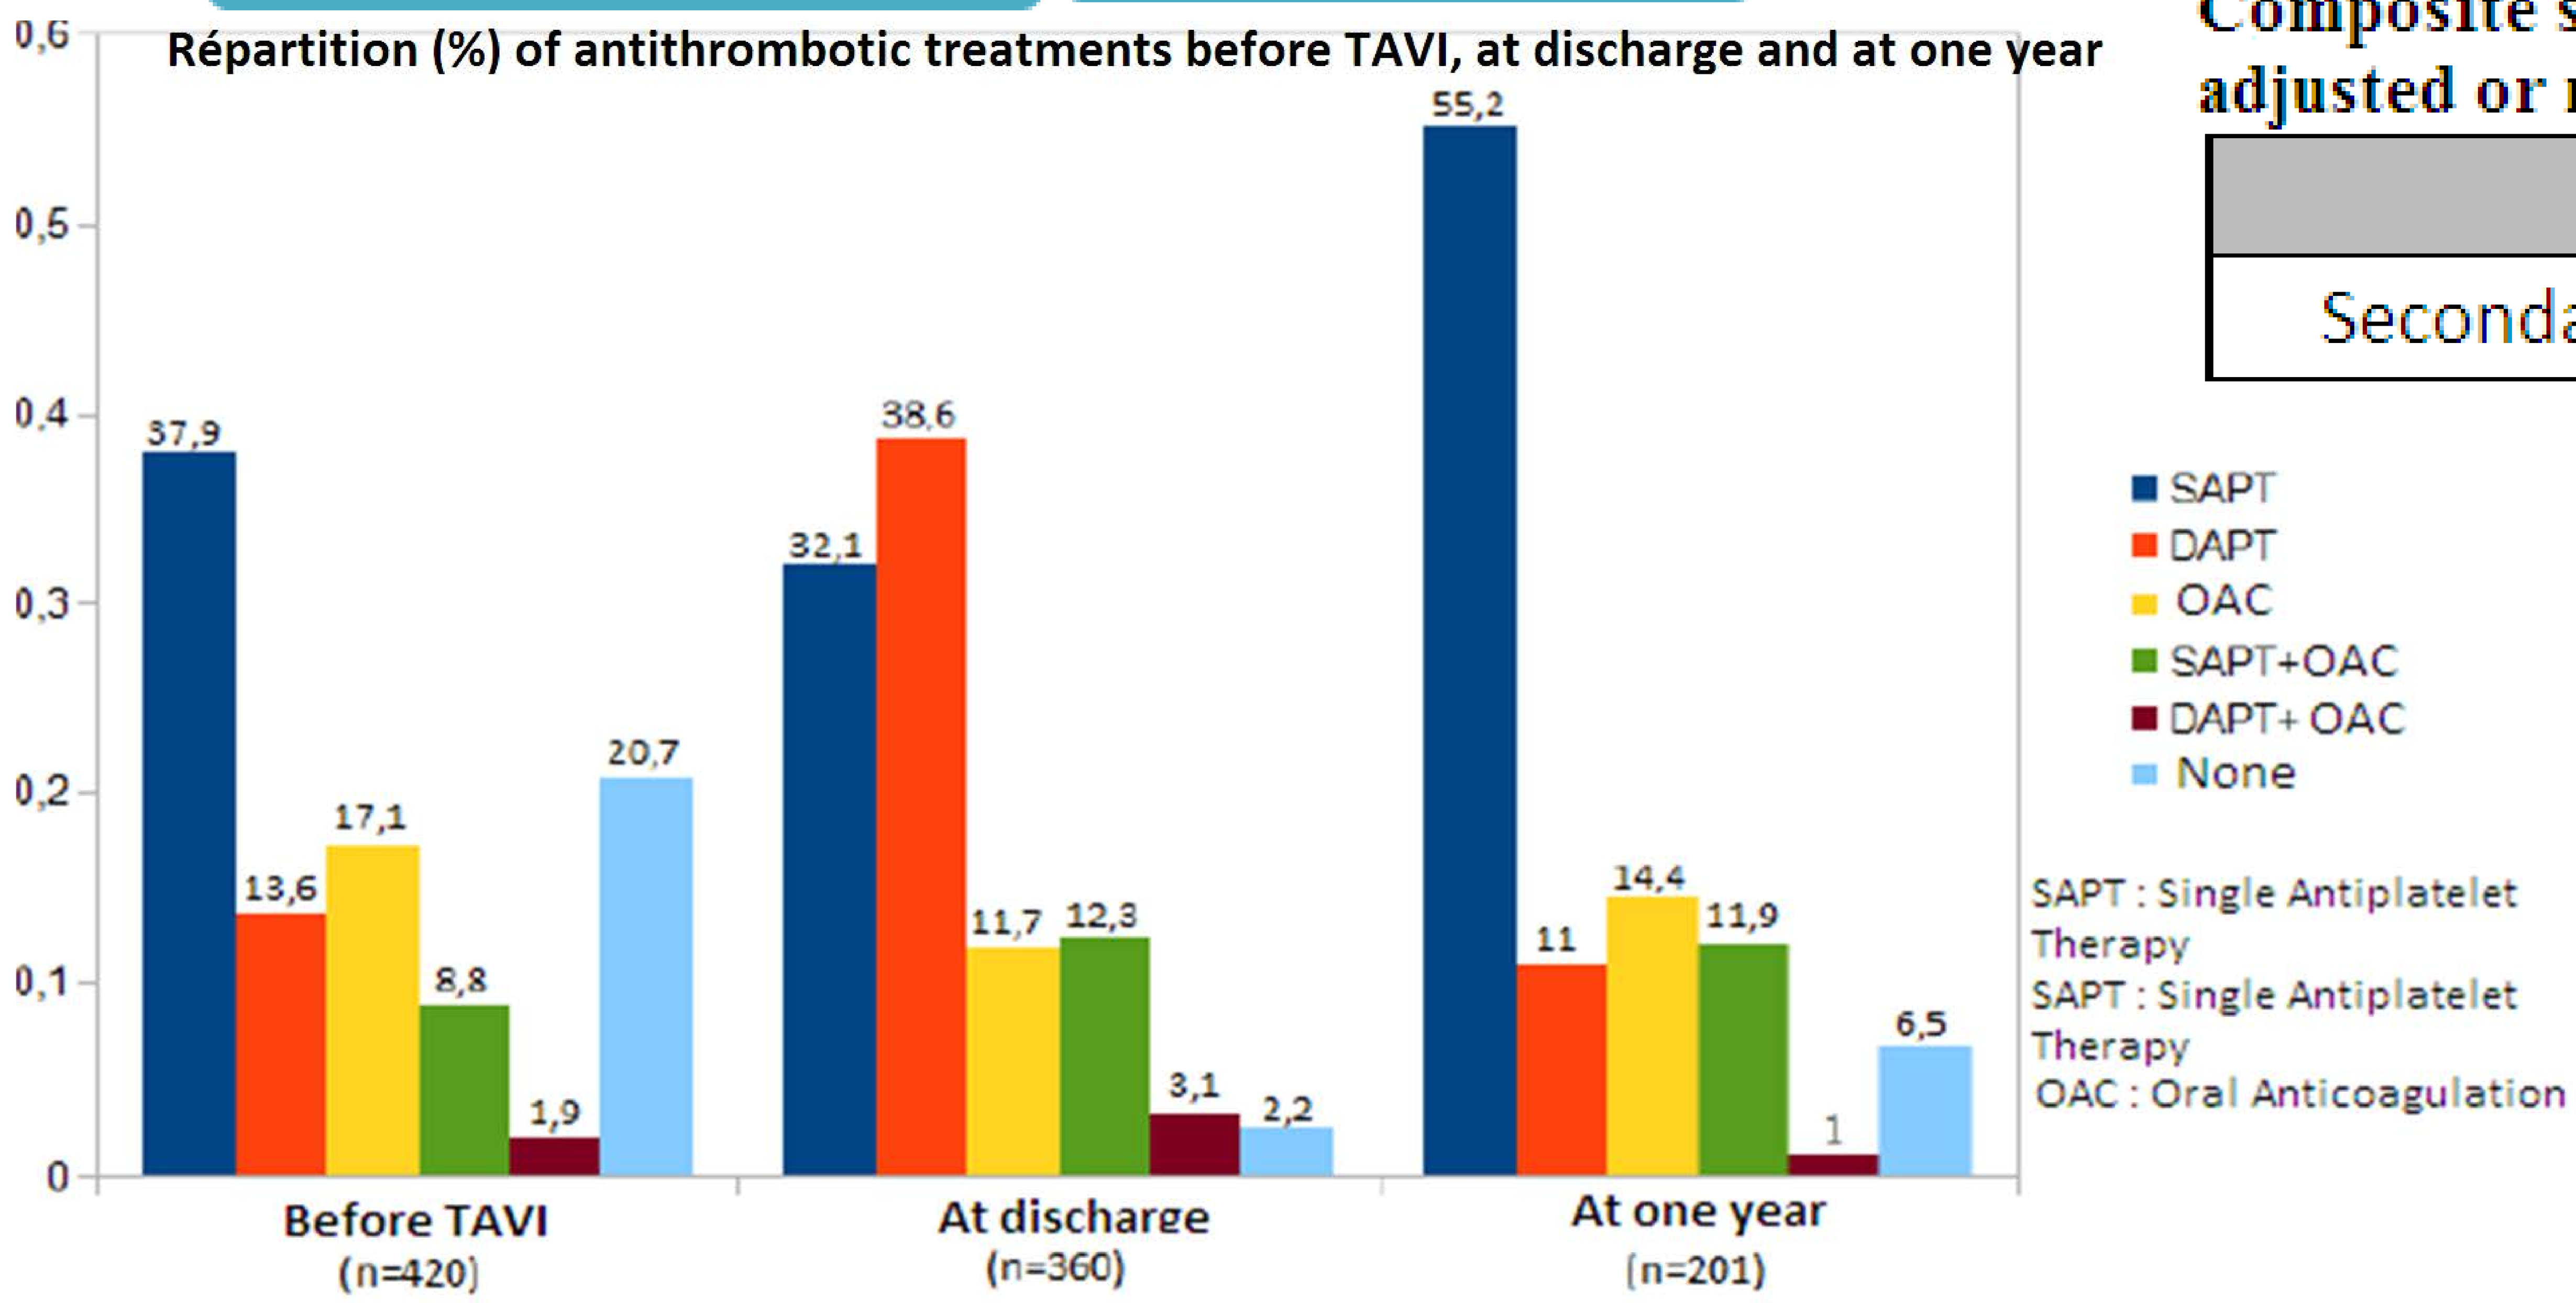
Répartition (%) of antithrombotic treatments before TAVI, at discharge and at one year
0,6
0,5
0,4
0,3
0,2
0,1
0
37,9
13,6
17,1
8,8
1,9
20,7
32,1
38,6
11,7
12,3
3,1
2,2
55,2
11
14,4
11,9
1
6,5
Before TAVI
(n=420)
At discharge
(n=360)
At one year
(n=201)
SAPT
DAPT
OAC
SAPT+OAC
DAPT+ OAC
None
SAPT : Single Antiplatelet Therapy
SAPT : Single Antiplatelet Therapy
OAC : Oral Anticoagulation

Composite primary endpoint according to VARC-2 in all patients and in non OAC and OAC groups

	All (n=360)	Non OAC (n=263)	OAC (n=97)	p
Primary endpoint n(%)	75 (20.8)	59 (22.4)	16 (16.5)	0.2183
After adjustment with propensity score	All (n=334)	Non OAC (n=243)	OAC (n=91)	p
Primary endpoint n(%)	69 (20,7)	53 (21.8)	16 (17.6)	0.3954

Composite secondary endpoint according to VARC-2 in all patients and in non OAC and OAC groups adjusted or not (same numbers)

	All (n=301)	Non OAC (n=263)	OAC (n=97)	p
Secondary endpoint n(%)	78 (25,9)	51 (23)	27 (34.2)	0.0510

- The rate of major bleeding did not differ although there was a trend for more bleeds in the OAC group (4% versus 2% in OAC and non-OAC groups, p=0.99).
- Coronary artery disease was the only independent correlate associated with the primary endpoint (OR=2.07, 95% CI [1.31-3.27])
- Atrial fibrillation and aortic valve area were independent correlates associated with the secondary endpoint (OR=2.71, IC95%[1.50 ;4.88] and OR=5.41, IC95%[1.27 ;23.00] respectively)

CONCLUSION

Antithrombotic regimen after TAVI deserves a randomized evaluation in this high-risk frail population.

The authors have no conflicts of interest