BIOMARKERS OF ACUTE KIDNEY INJURY IN ST-ELEVATED MYOCARDIAL INFARCTION TREATED BY PERCUTANEOUS CORONARY INTERVENTION

Lee Nguyen MD, Vincent Spagnoli MD, Mathieu Kerneis MD, Delphine Brugier, Nicolas Vignolles BS, Sophie Gallier BS, Jean-Philippe Collet MD-PhD, Gilles Montalescot MD-PhD, and Johanne Silvain MD-PhD

Université Paris 6 (UPMC), Hôpital de la Pitié-Salpêtrière (AP-HP), INSERM UMRS_1166, Paris , France.

Abstract

Background

Contrast induced acute kidney injury (CI-AKI) is associated with a high morbidity and mortality in coronary patients. We evaluated the predictive value of the biomarkers of renal dysfunction, Cystatin-C and neutrophil gelatinase-associated lipocalin (NGAL) in the incidence of CI-AKI and their prognostic value on mortality in ST-segment elevated myocardial infarction (STEMI) patients treated by primary percutaneous coronary intervention (pPCI).

Methods

NGAL and cystatin-C level were measured prior to any contrast injection in 701 STEMI patients and were correlated to the occurrence of CI-AKI according to the various existing definitions. Association between biomarkers levels and the incidence of CI-AKI, MACE and all-cause mortality at 1-year-follow-up were evaluated. The add-on impact of biomarkers on top of clinical data was assessed on diagnosis and prognosis.

Results

Increasing levels of plasmatic NGAL and cystatin-C were associated with a stepwise increase in the incidence of AKI and with renal failure stage. Both biomarkers significantly predicted CI-AKI with an AUC of 0.60 and 0.62 for cystatin-C, and for NGAL, respectively ($p < 0.05$). MACE and all-cause mortality at 1-year-follow-up were also higher in the third tertile of both biomarkers (p value <0.01). After adjustment, higher levels of NGAL were associated with the incidence CI-AKI (adjOR 1.7 (CI 1.01-2.90) p=0.049) but not with mortality; while cystatin-C was not associated with CI-AKI or mortality.

Fundings

Grant to Dr Joahnne Silvain from the Société Française de Cardiolgoie (2012) .
Action Coeur study group ([www.actioncoeur.fr)](http://www.actioncoeur.fr)

Biomarkers and relationship with CI-AKI, MACE and mortality

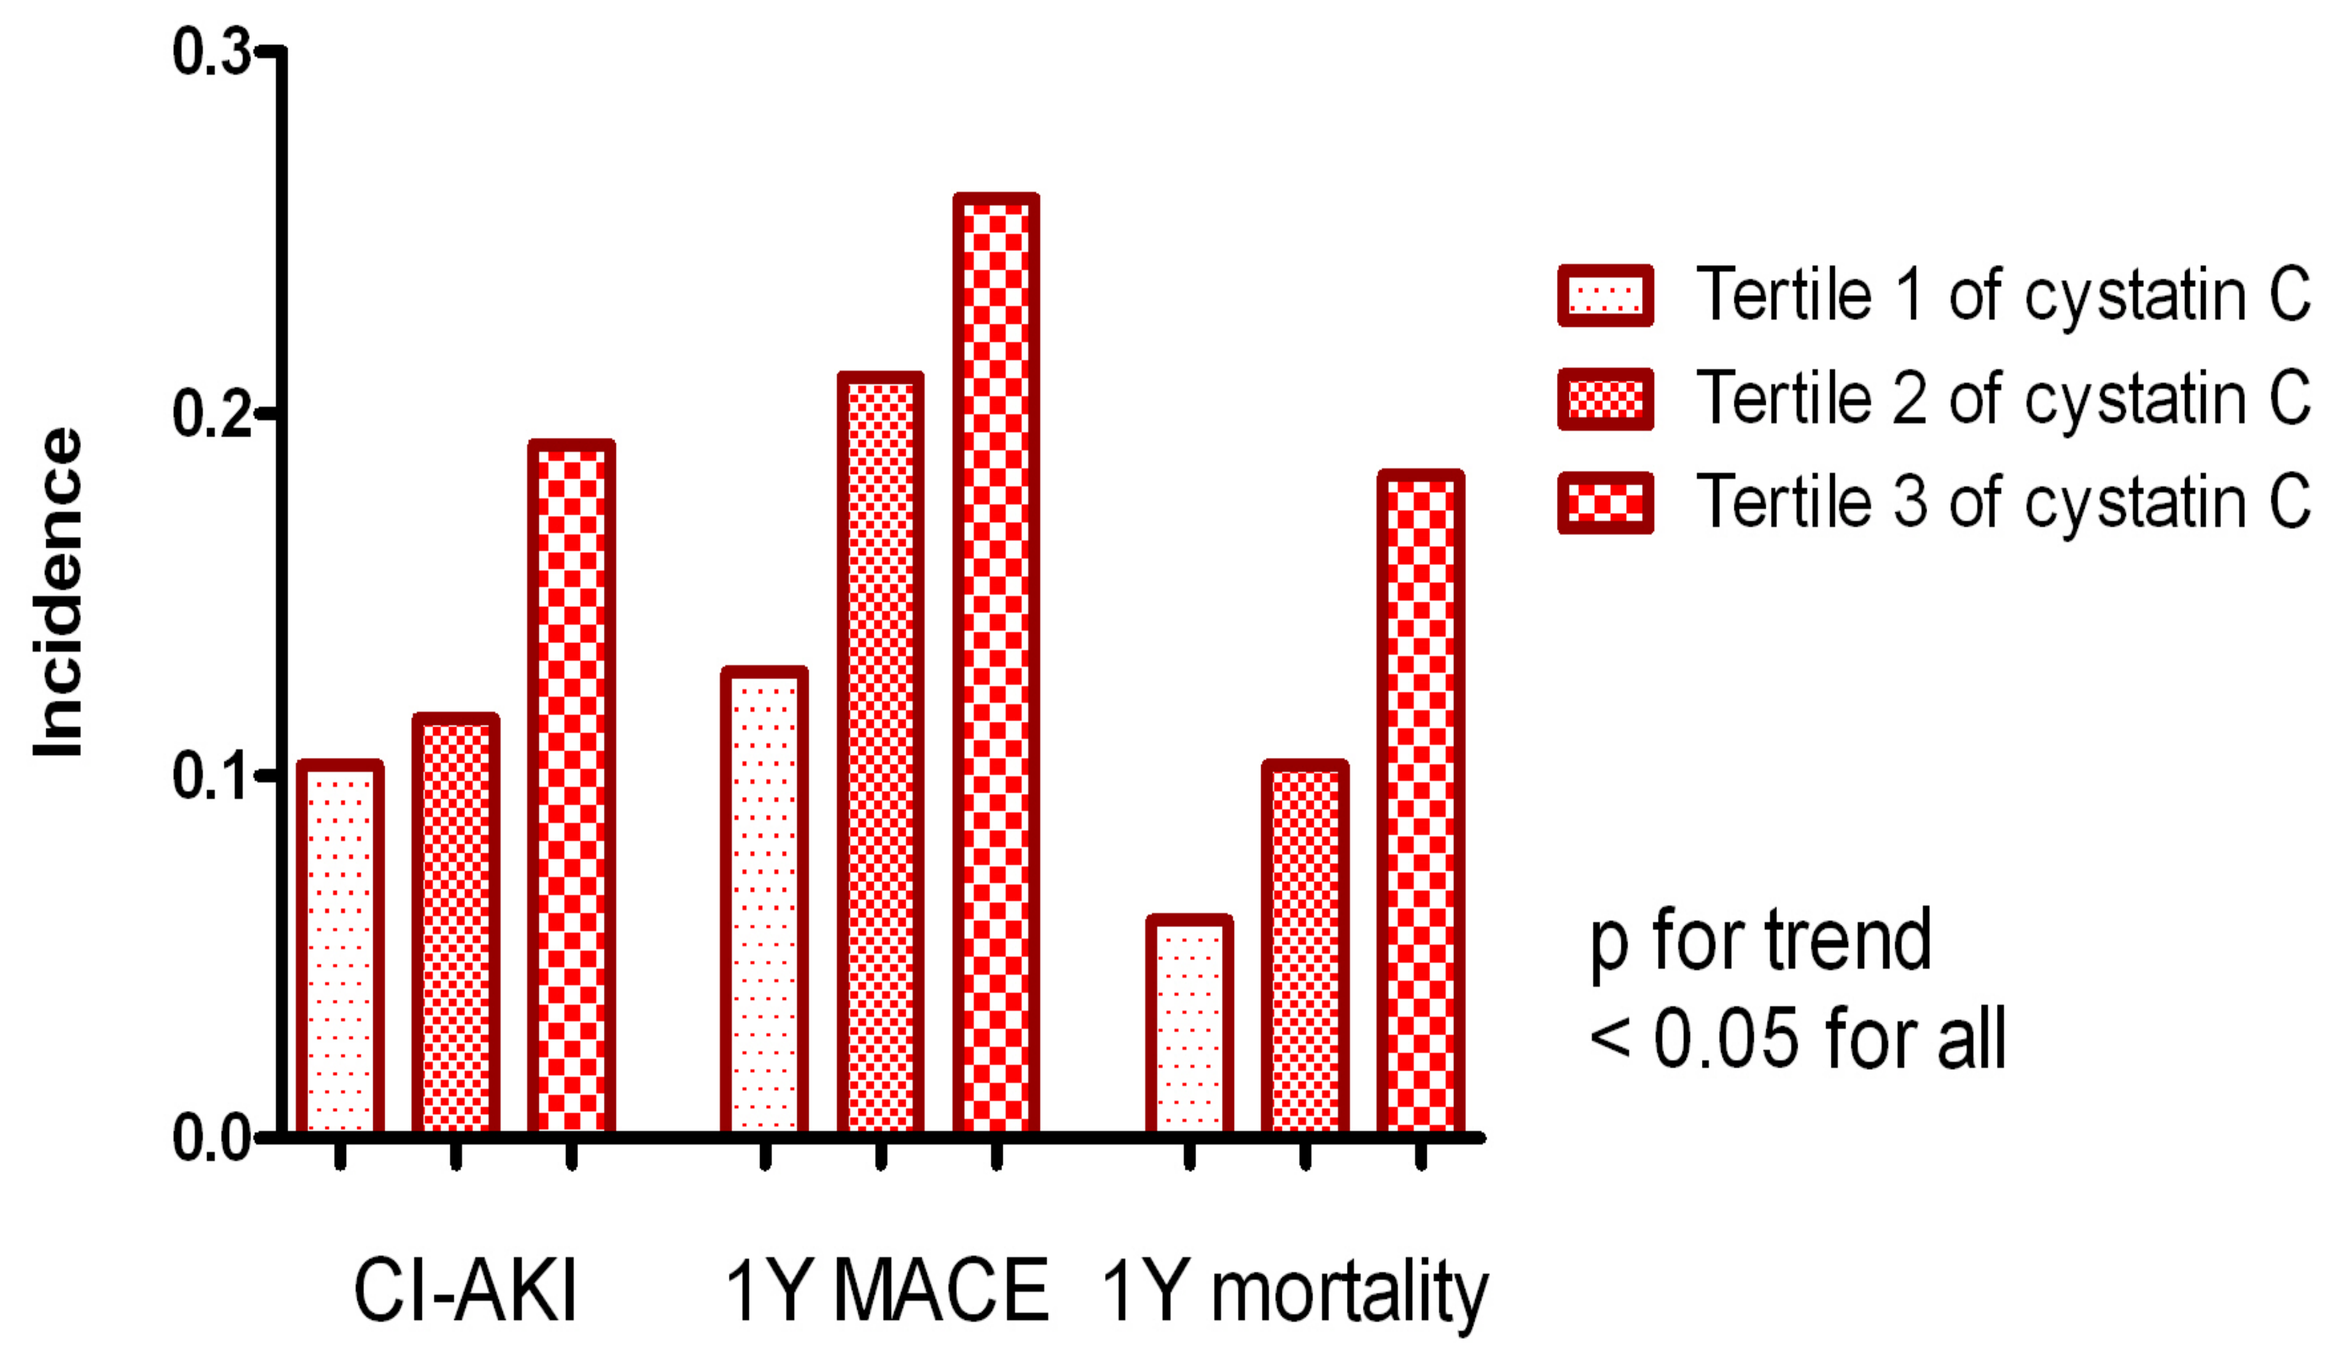
Incidence
0.0
0.1
0.2
0.3
Tertile 1 of cystatin C
Tertile 2 of cystatin C
Tertile 3 of cystatin C
p for trend
< 0.05 for all
CI-AKI
1Y MACE
1Y mortality

Figure 1. Events according to cystatin C tertile

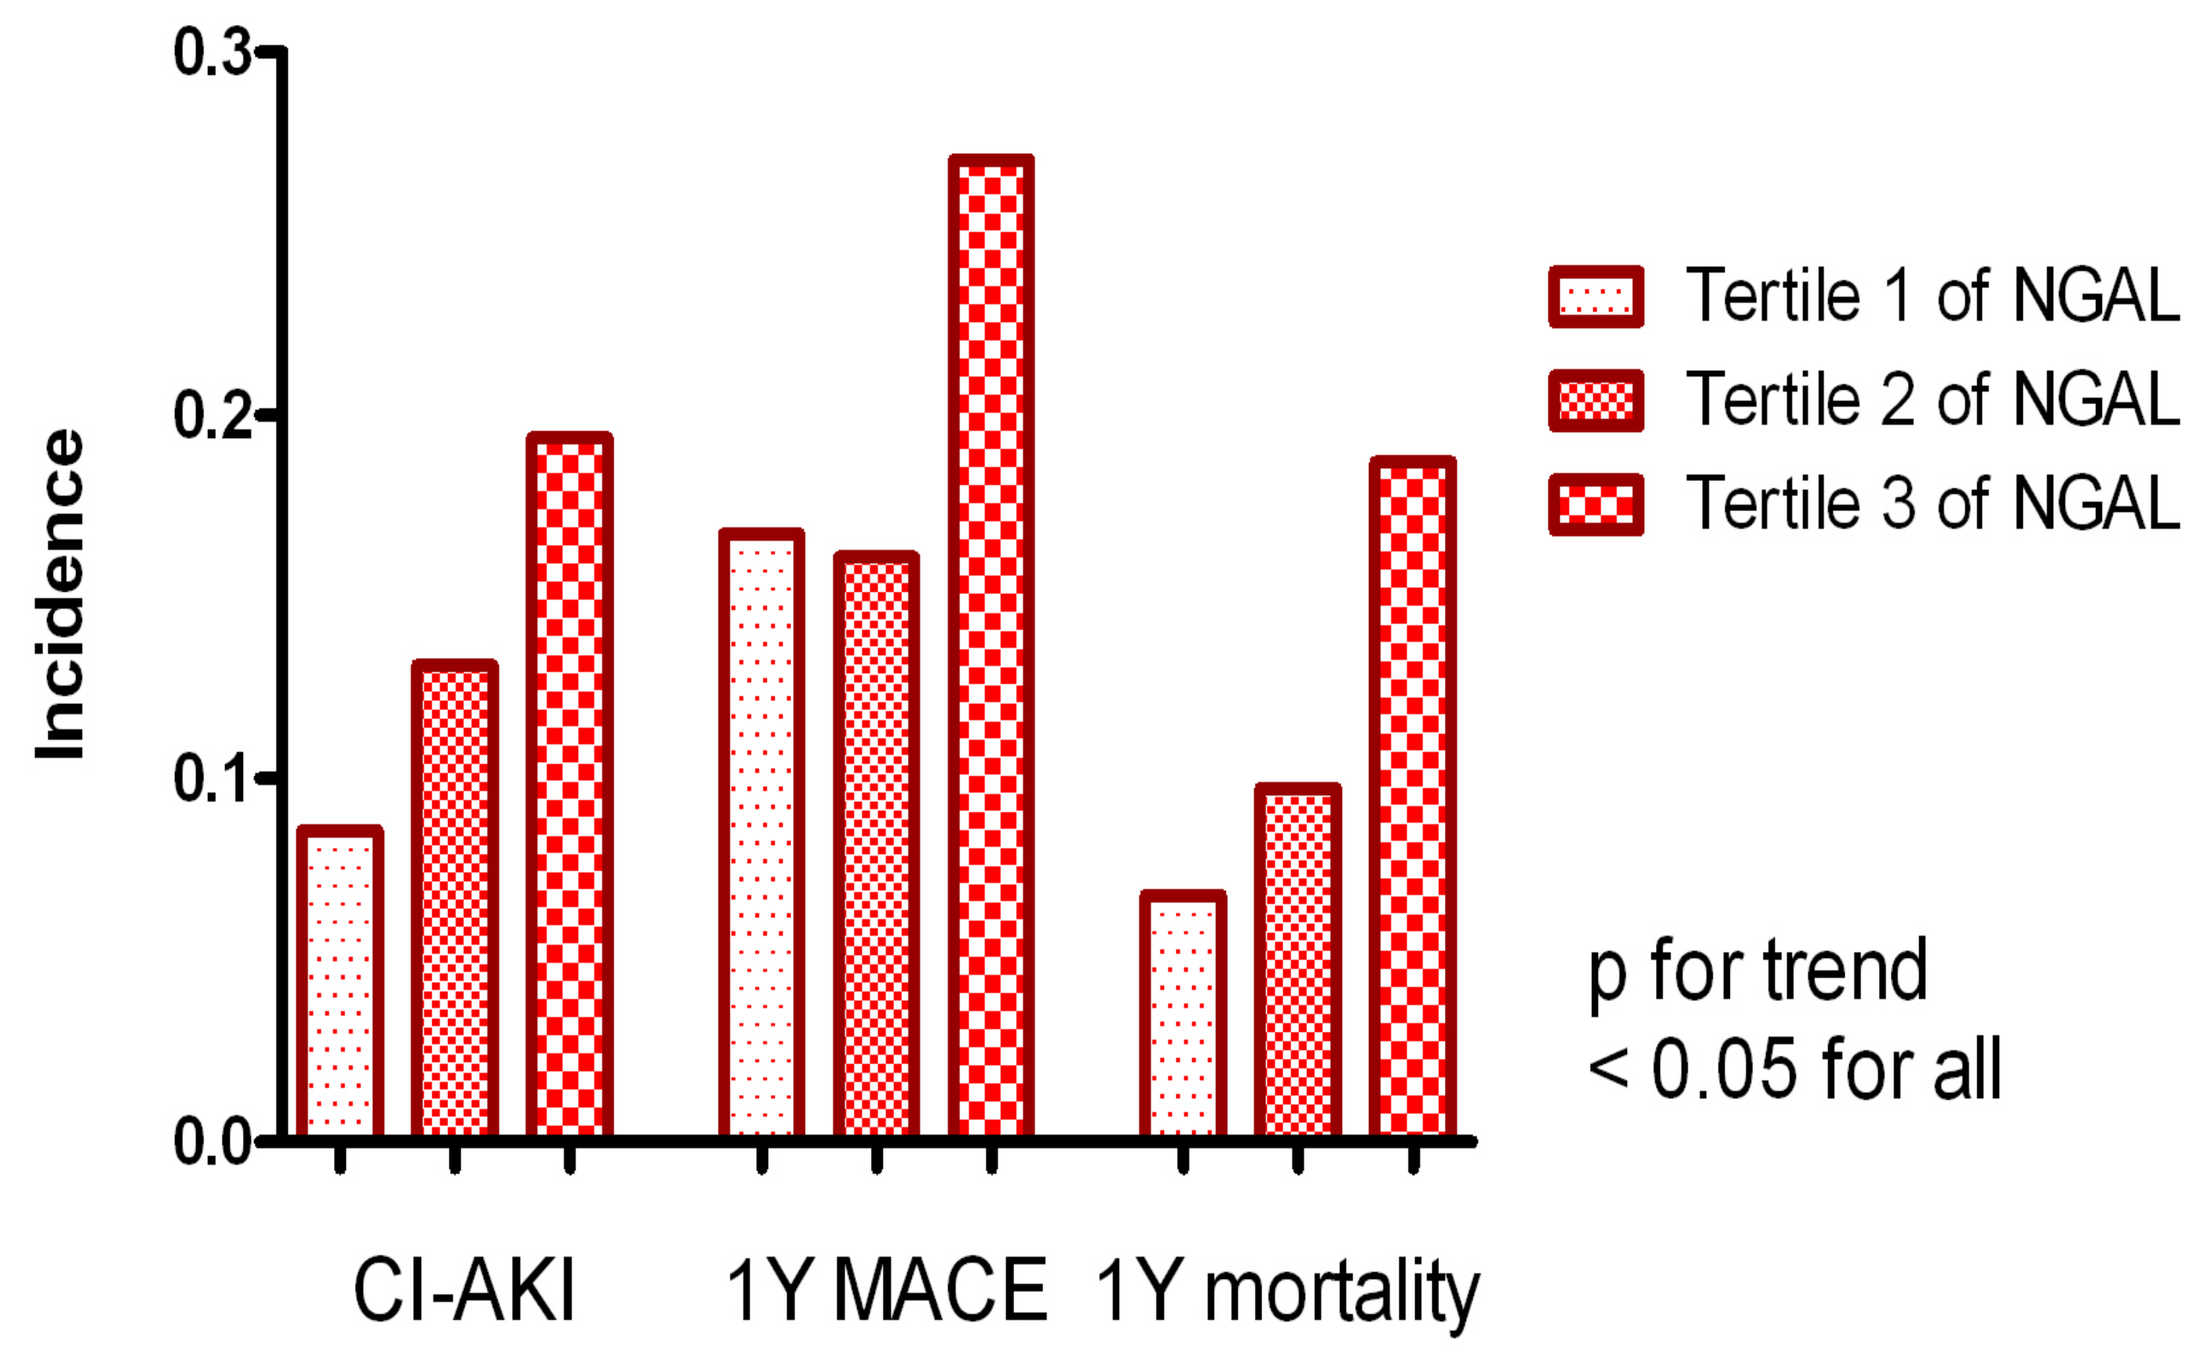
Incidence
0.0
0.1
0.2
0.3
Tertile 1 of NGAL
Tertile 2 of NGAL
Tertile 3 of NGAL
p for trend
< 0.05 for all
CI-AKI
1Y MACE
1Y mortality

Figure 2. Events according to NGAL tertile

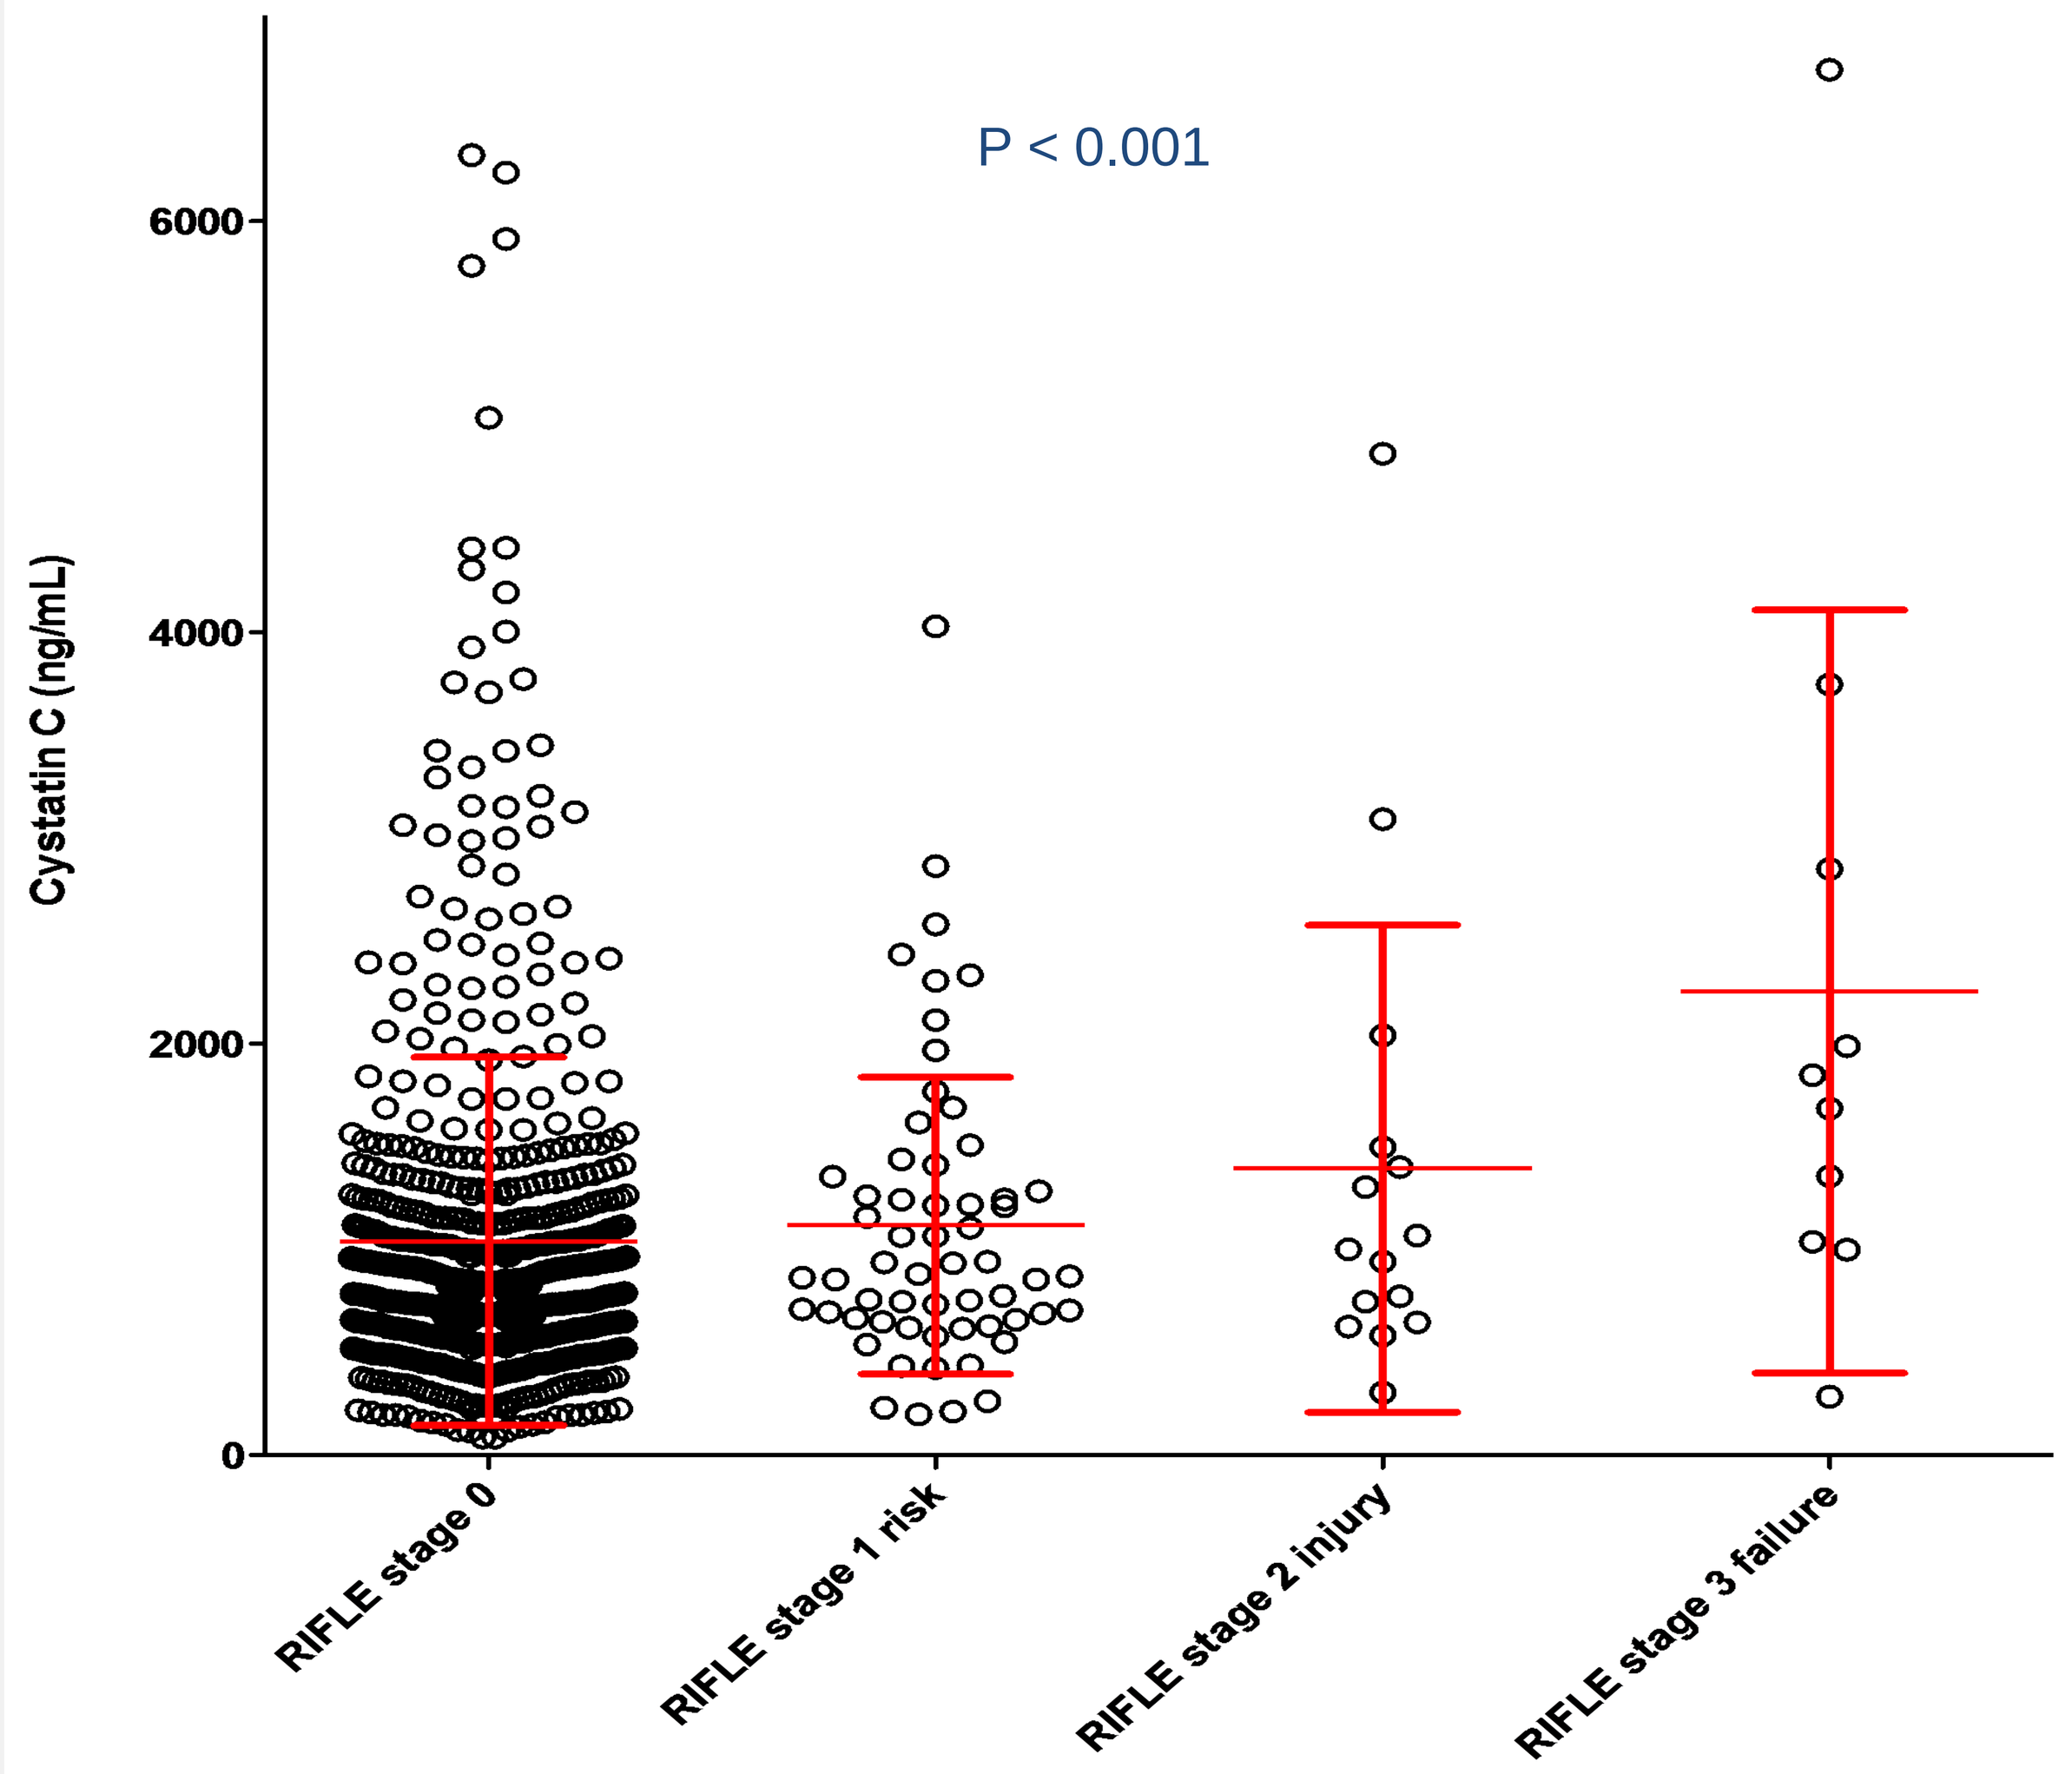
P < 0.001
Cystatin C (ng/mL)
0
2000
4000
6000
RIFLE stage 0
RIFLE stage 1 risk
RIFLE stage 2 injury
RIFLE stage 3 failure

Figure 3. Severity of CI-AKI according to Cystatin C level

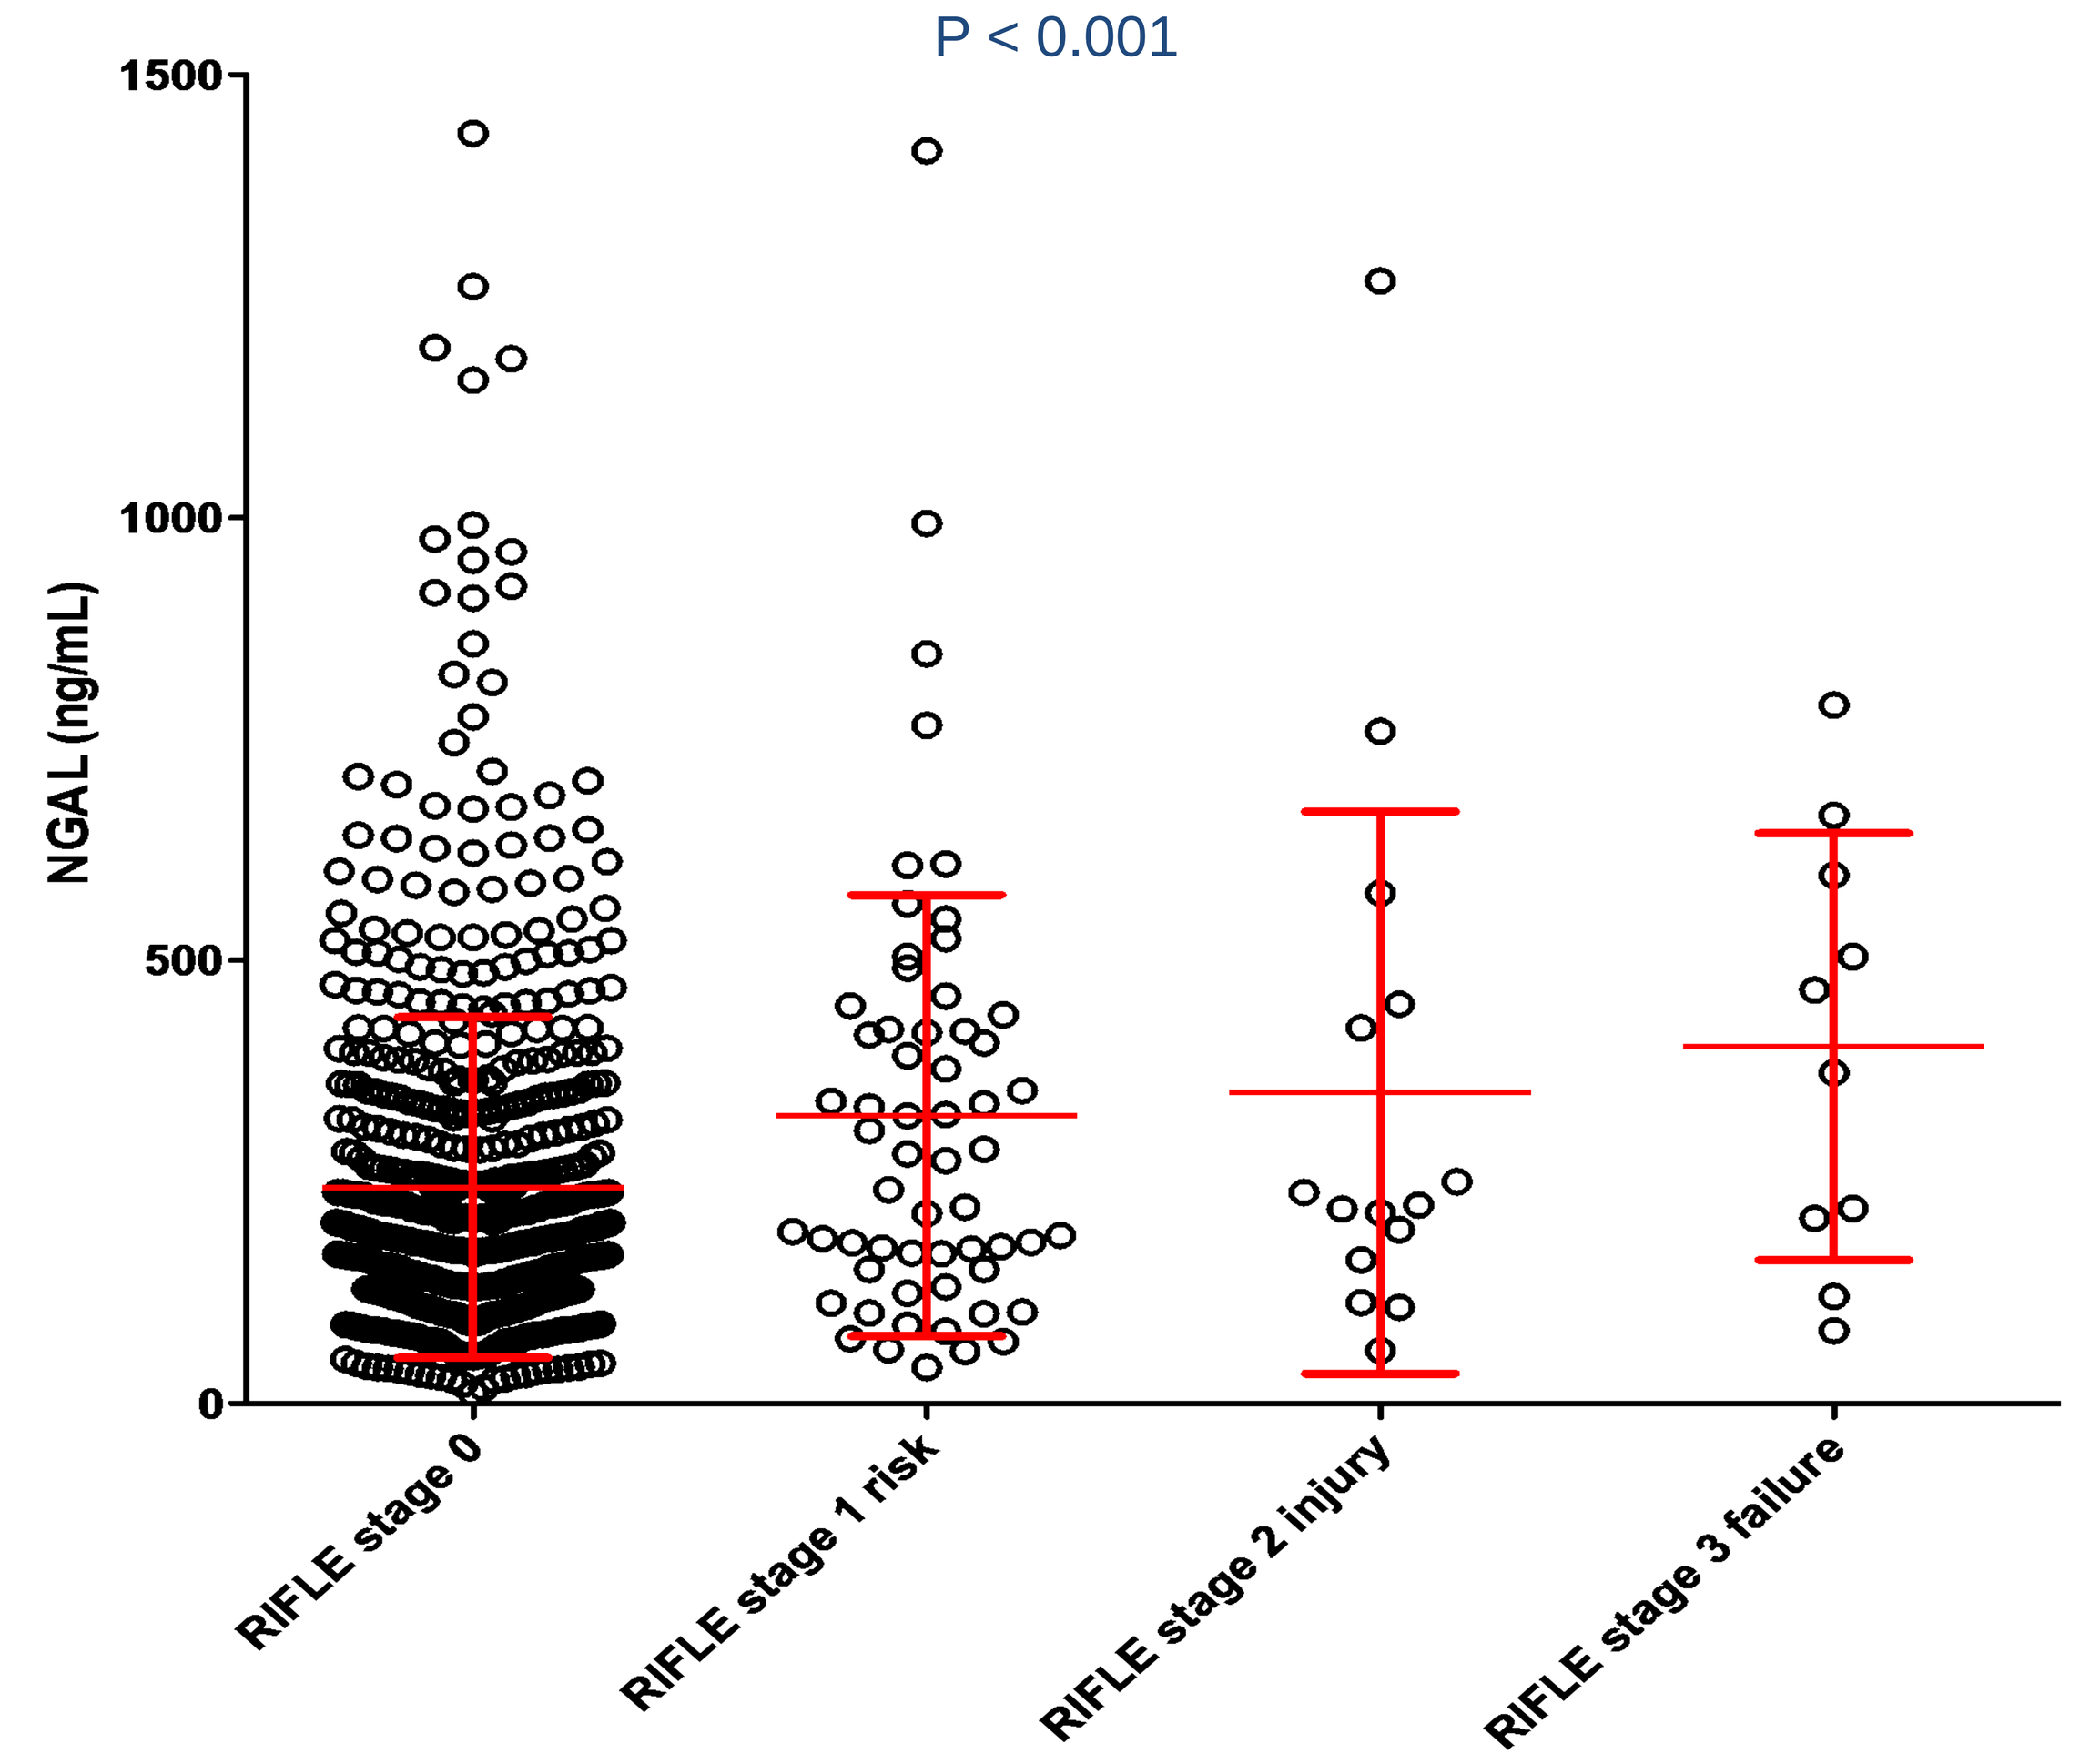
P < 0.001
NGAL (ng/mL)
0
500
1000
1500
RIFLE stage 0
RIFLE stage 1 risk
RIFLE stage 2 injury
RIFLE stage 3 failure

Figure 4. Severity of CI-AKI according to NGAL level

Overall prognosis value of NGAL and cystatin C in STEMI

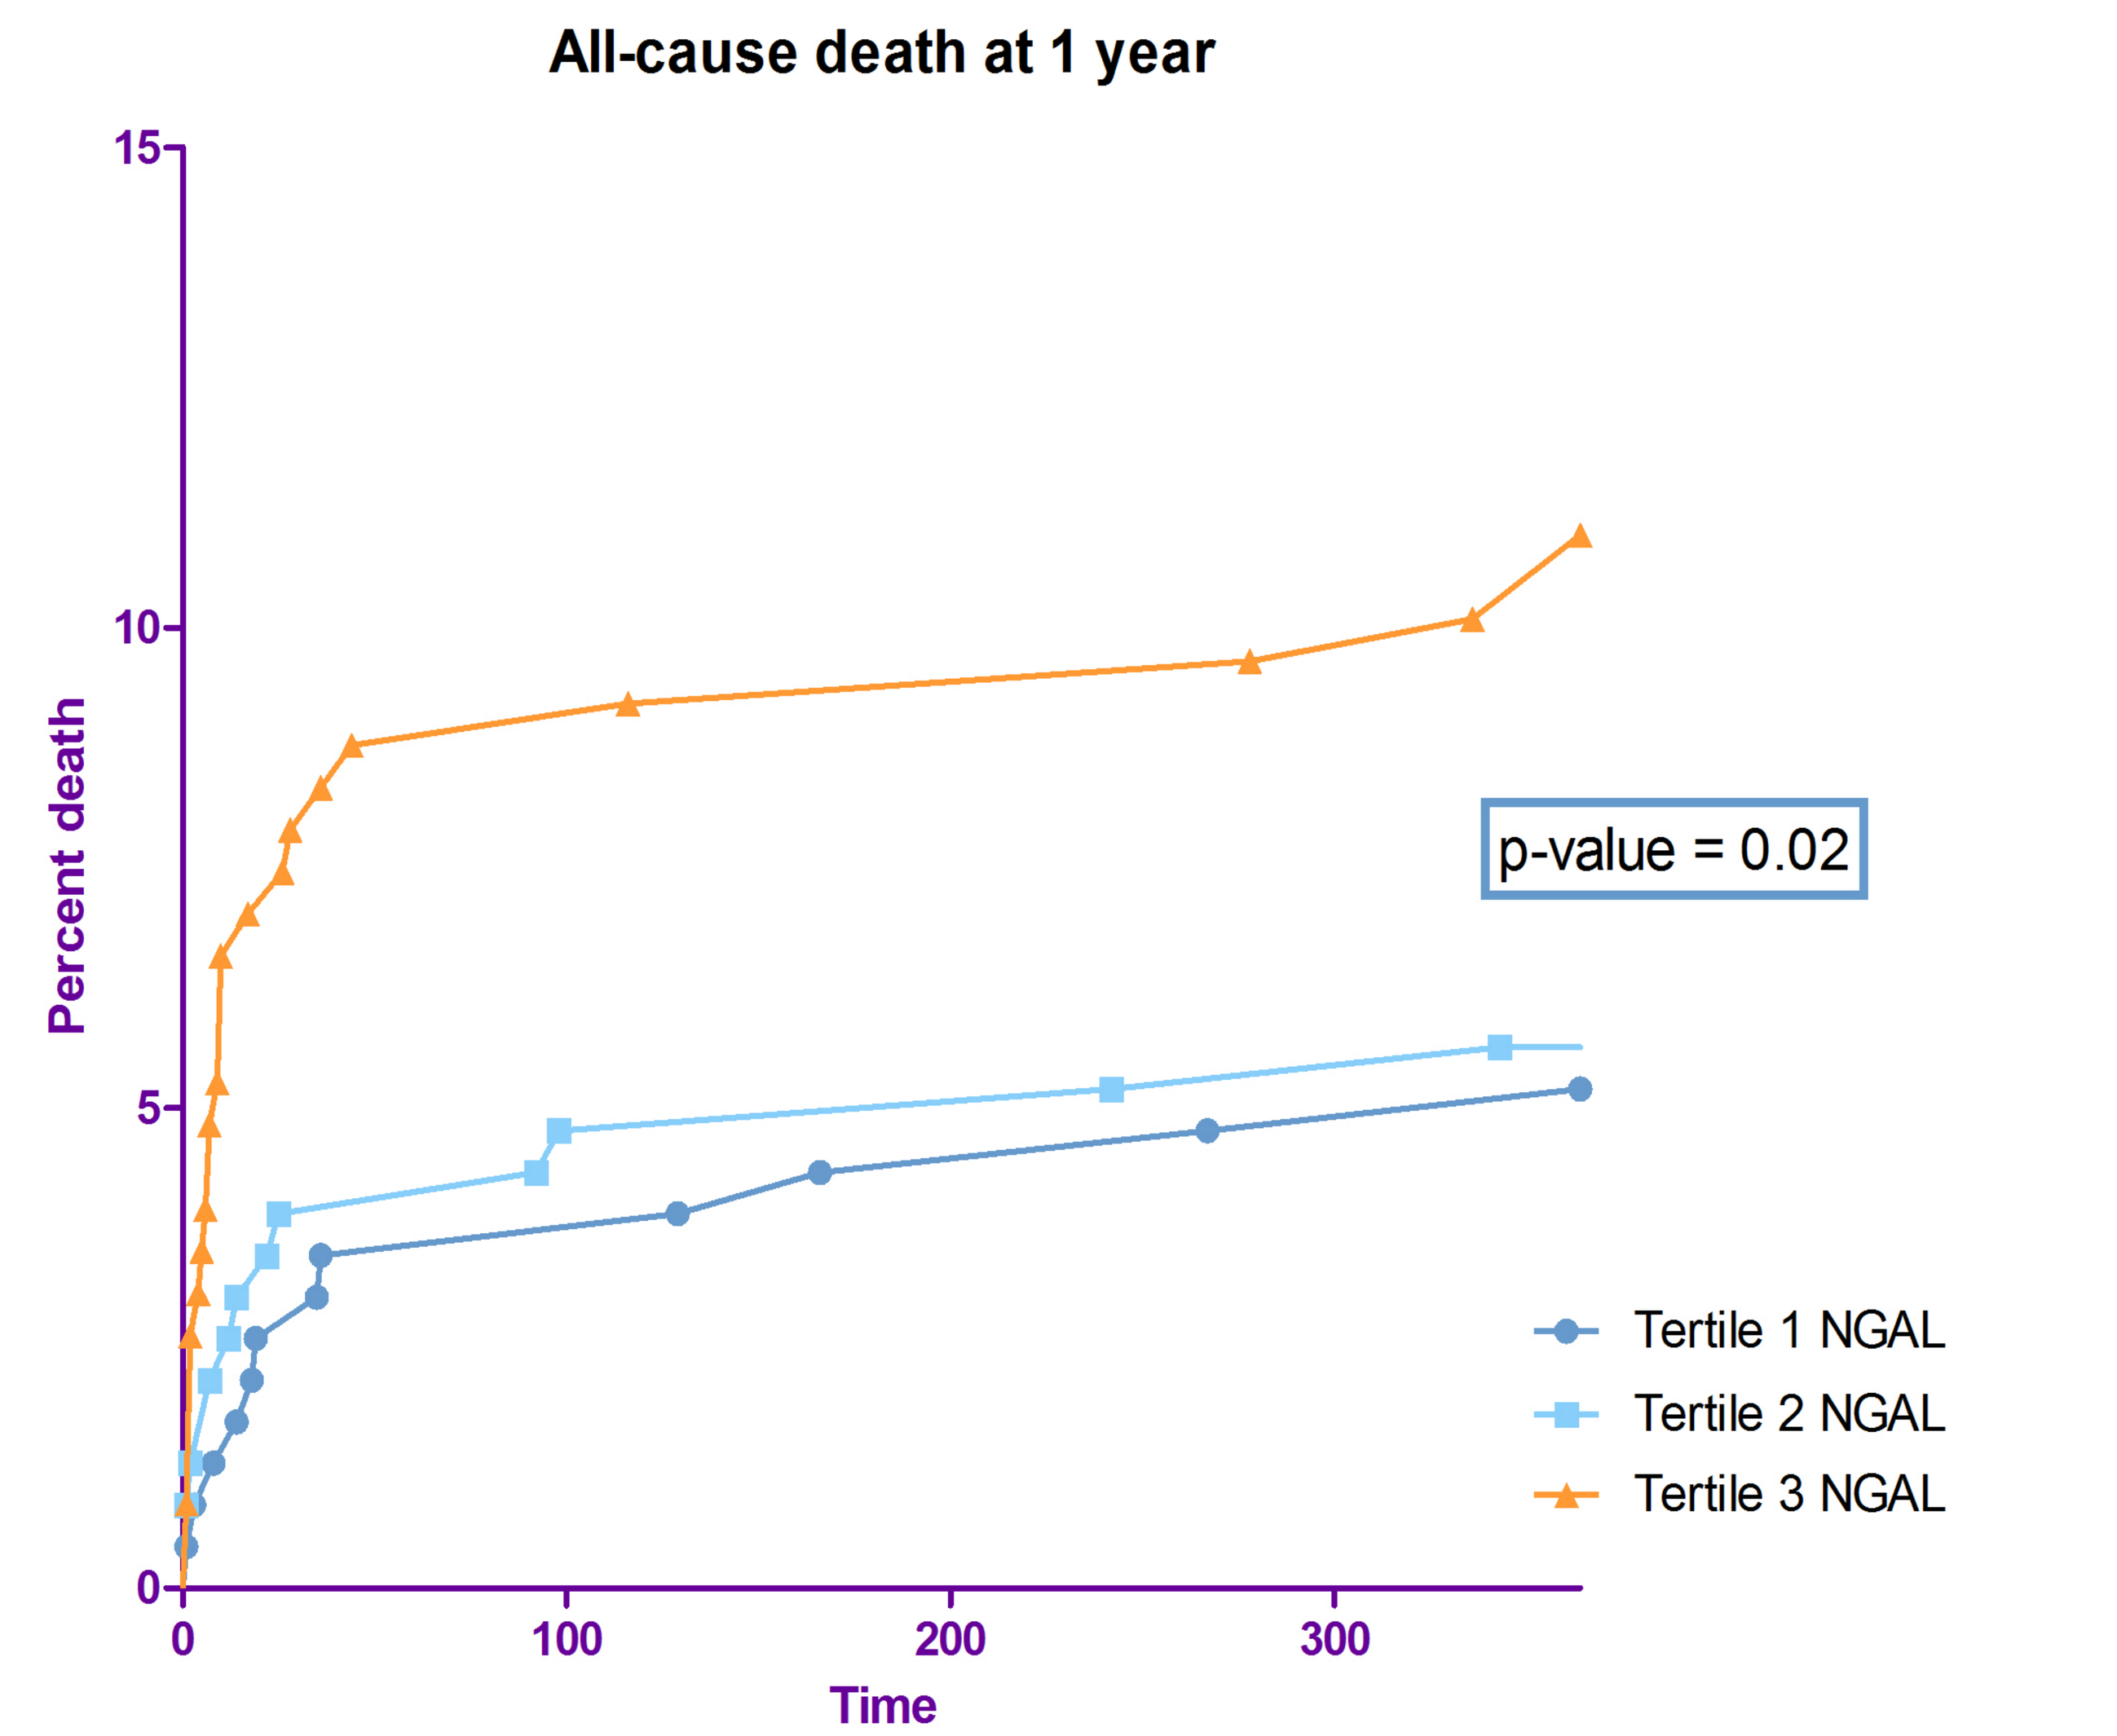
All-cause death at 1 year
Percent death
0
5
10
15
p-value = 0.02
Tertile 1 NGAL
Tertile 2 NGAL
Tertile 3 NGAL
0
100
200
300
Time

Figure 5. Survival curves for all-cause death at 1 year according to NGAL tertile

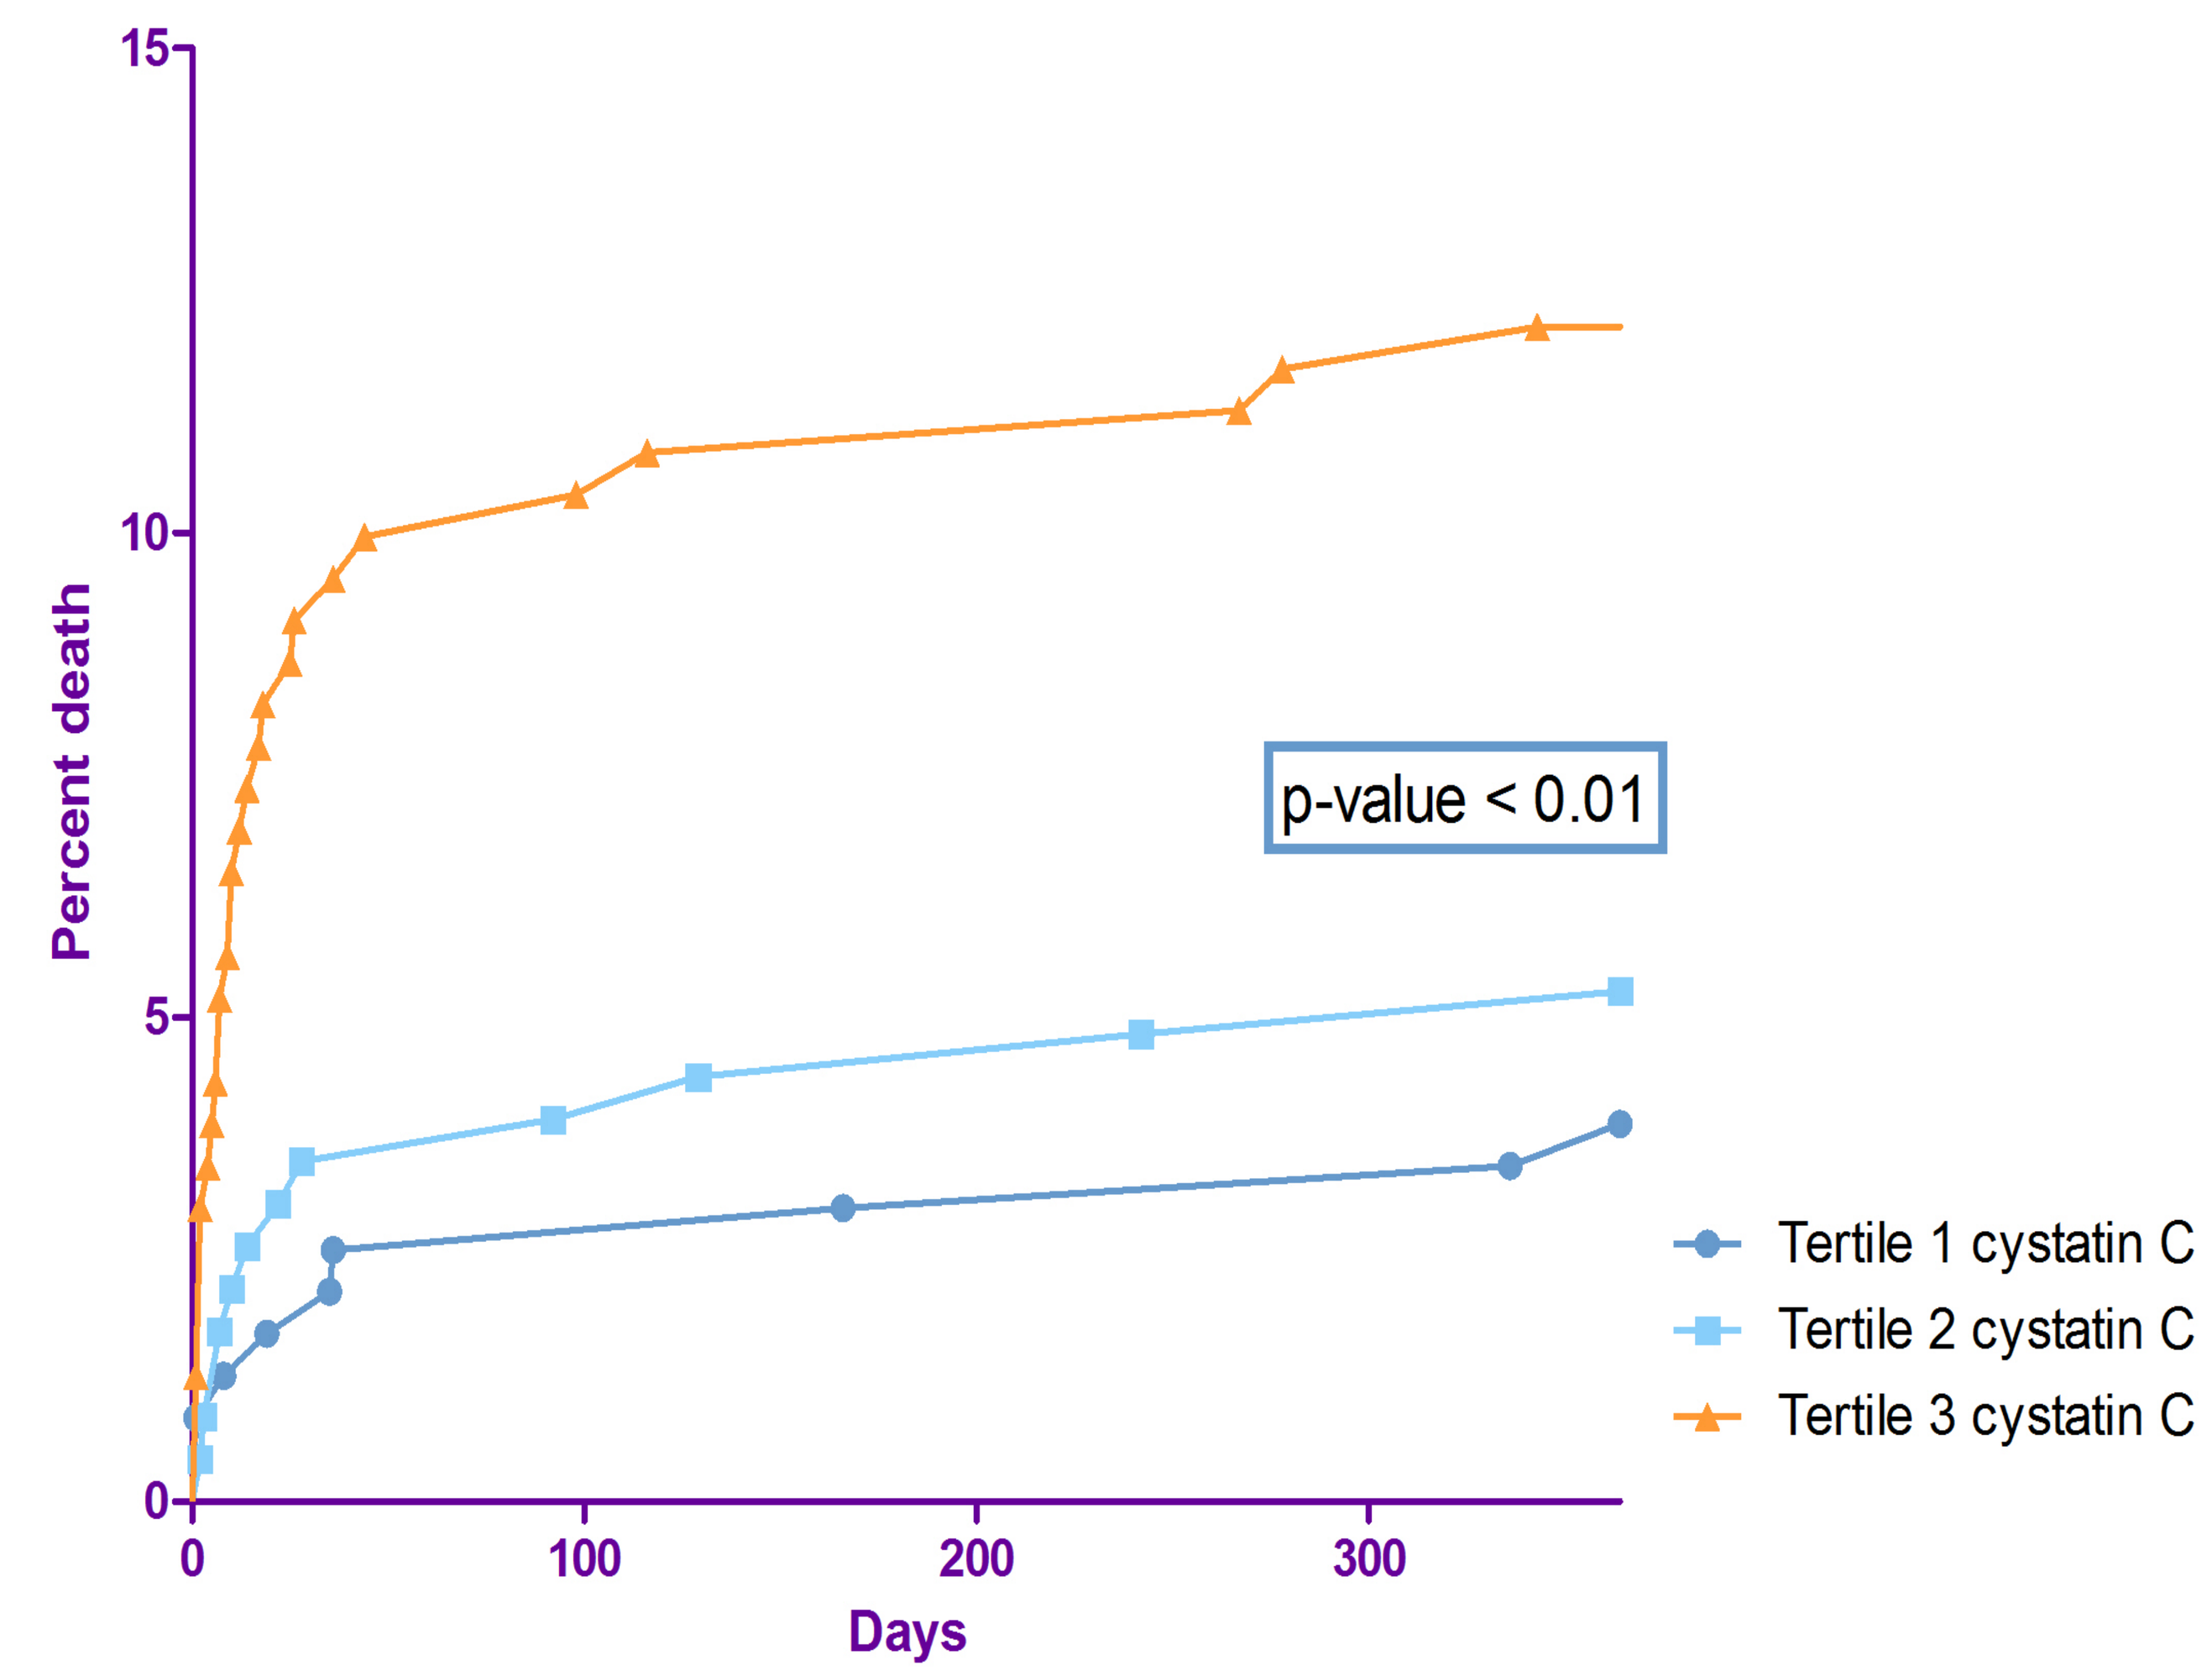
Percent death
0
5
10
15
p-value < 0.01
Tertile 1 cystatin C
Tertile 2 cystatin C
Tertile 3 cystatin C
0
100
200
300
Days

Figure 6. Survival curves for all-cause death at 1 year according to cystatin C tertile

Société Française de Cardiologie

Conclusion

Although NGAL and cystatin C are associated with the incidence and severity of CI-AKI and also with MACE and 1-year mortality ; they do not provide additive value as compared to baseline clinical risk factors in STEMI patients treated by primary PCI

INSTITUT DE CARDIOLOGIE
Groupe Hospitalier Pitié-Salpêtrière