Is left ventricular longitudinal strain a good pronostic factor in friedreich ataxia?

P903

C.Heuzé[1], L. Legrand[1], A. Diallo[2], M-L. Monin[3], C. Ewenczyk[3], R. Isnard[1,2], E. Vicaut[2], A. Durr[3], F. Pousset[1]

[1]Sorbonne University , Cardiology Department, APHP, Pitié-Salpêtrière University Hospital Paris, France [2] ACTION (Allies in Cardiovascular Trials Initiatives and Organized Networks) Academic organization, Sorbonne University, Paris, France [3]Genetic Department, Brain & Spine Institute , APHP, Pitié-Salpêtrière University Hospital, Paris, France.

INTRODUCTION

Friedreich Ataxia (FRDA) is due to mitochondrial dysfunction caused by abnormal repetition of GAA expansion:

- First genetic cerebellar ataxia: 1/50000 patients
- Autosomal recessive transmission
- Moderate cardiac hypertrophy: frequent abnormal ECG (80%)
- Cardiopathy is the main cause of death in FRDA before 40 years old (heart failure)
- Cardiological evolution is slow with a progressive decrease in Left Ventricular Ejection Fraction (LVEF)
- Global Longitudinal strain is an effective technique to detect subtitles changes in LV function

OBJECTIVES: To evaluate the prognostic value of global longitudinal strain (GLS) in patient with FRDA as compared to LVEF.

RESULTS (1): Clinical biological and echocardiographic characteristics of the population

a) Clinical data

Male: 51%, mean age = 35,1 ± 12y
Age at diagnostic: 17,2 ± 10,6y
Wheelchair patients: 72% aged 26 ± 10 y
Symptoms (20%): 50% dyspnea, 26% palpitations chest pain 24%

b) ECG data

Abnormal ECG: n=136 (87%)
BBD n=31 (20%)
Negatives T wave :n=126 (80%)

c) Echocardiographic data

SWT (mm): 11,4 ± 2,5 LVEDD (mm):44,4 ± 5,5
PP (mm): 10,4 ± 1,8 LVESD (mm): 27 ± 6,3
LVMI (g/m²): 99±26
OG area (cm²): 15,3 ± 4,2 , RWT: 0,5 ± 0,1

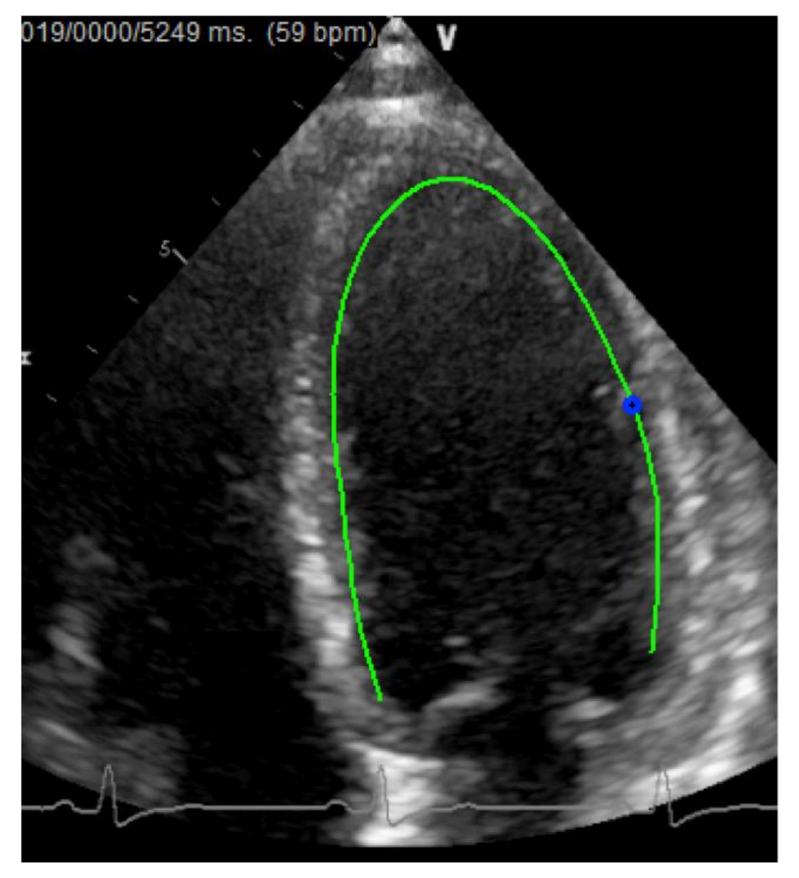

LVEF and GLS

	population	alive
LVEF (%) N=156	**63.5 ± 8.6**	**64.7 ± 6.2**
GLS (%), N=141	**- 19.9 ± 5.0**	**- 20.5 ± 4.4**

Correlation coefficient: GLS and LVEF: r = 0.31 (0.15 ; 0.45), p=0.0002

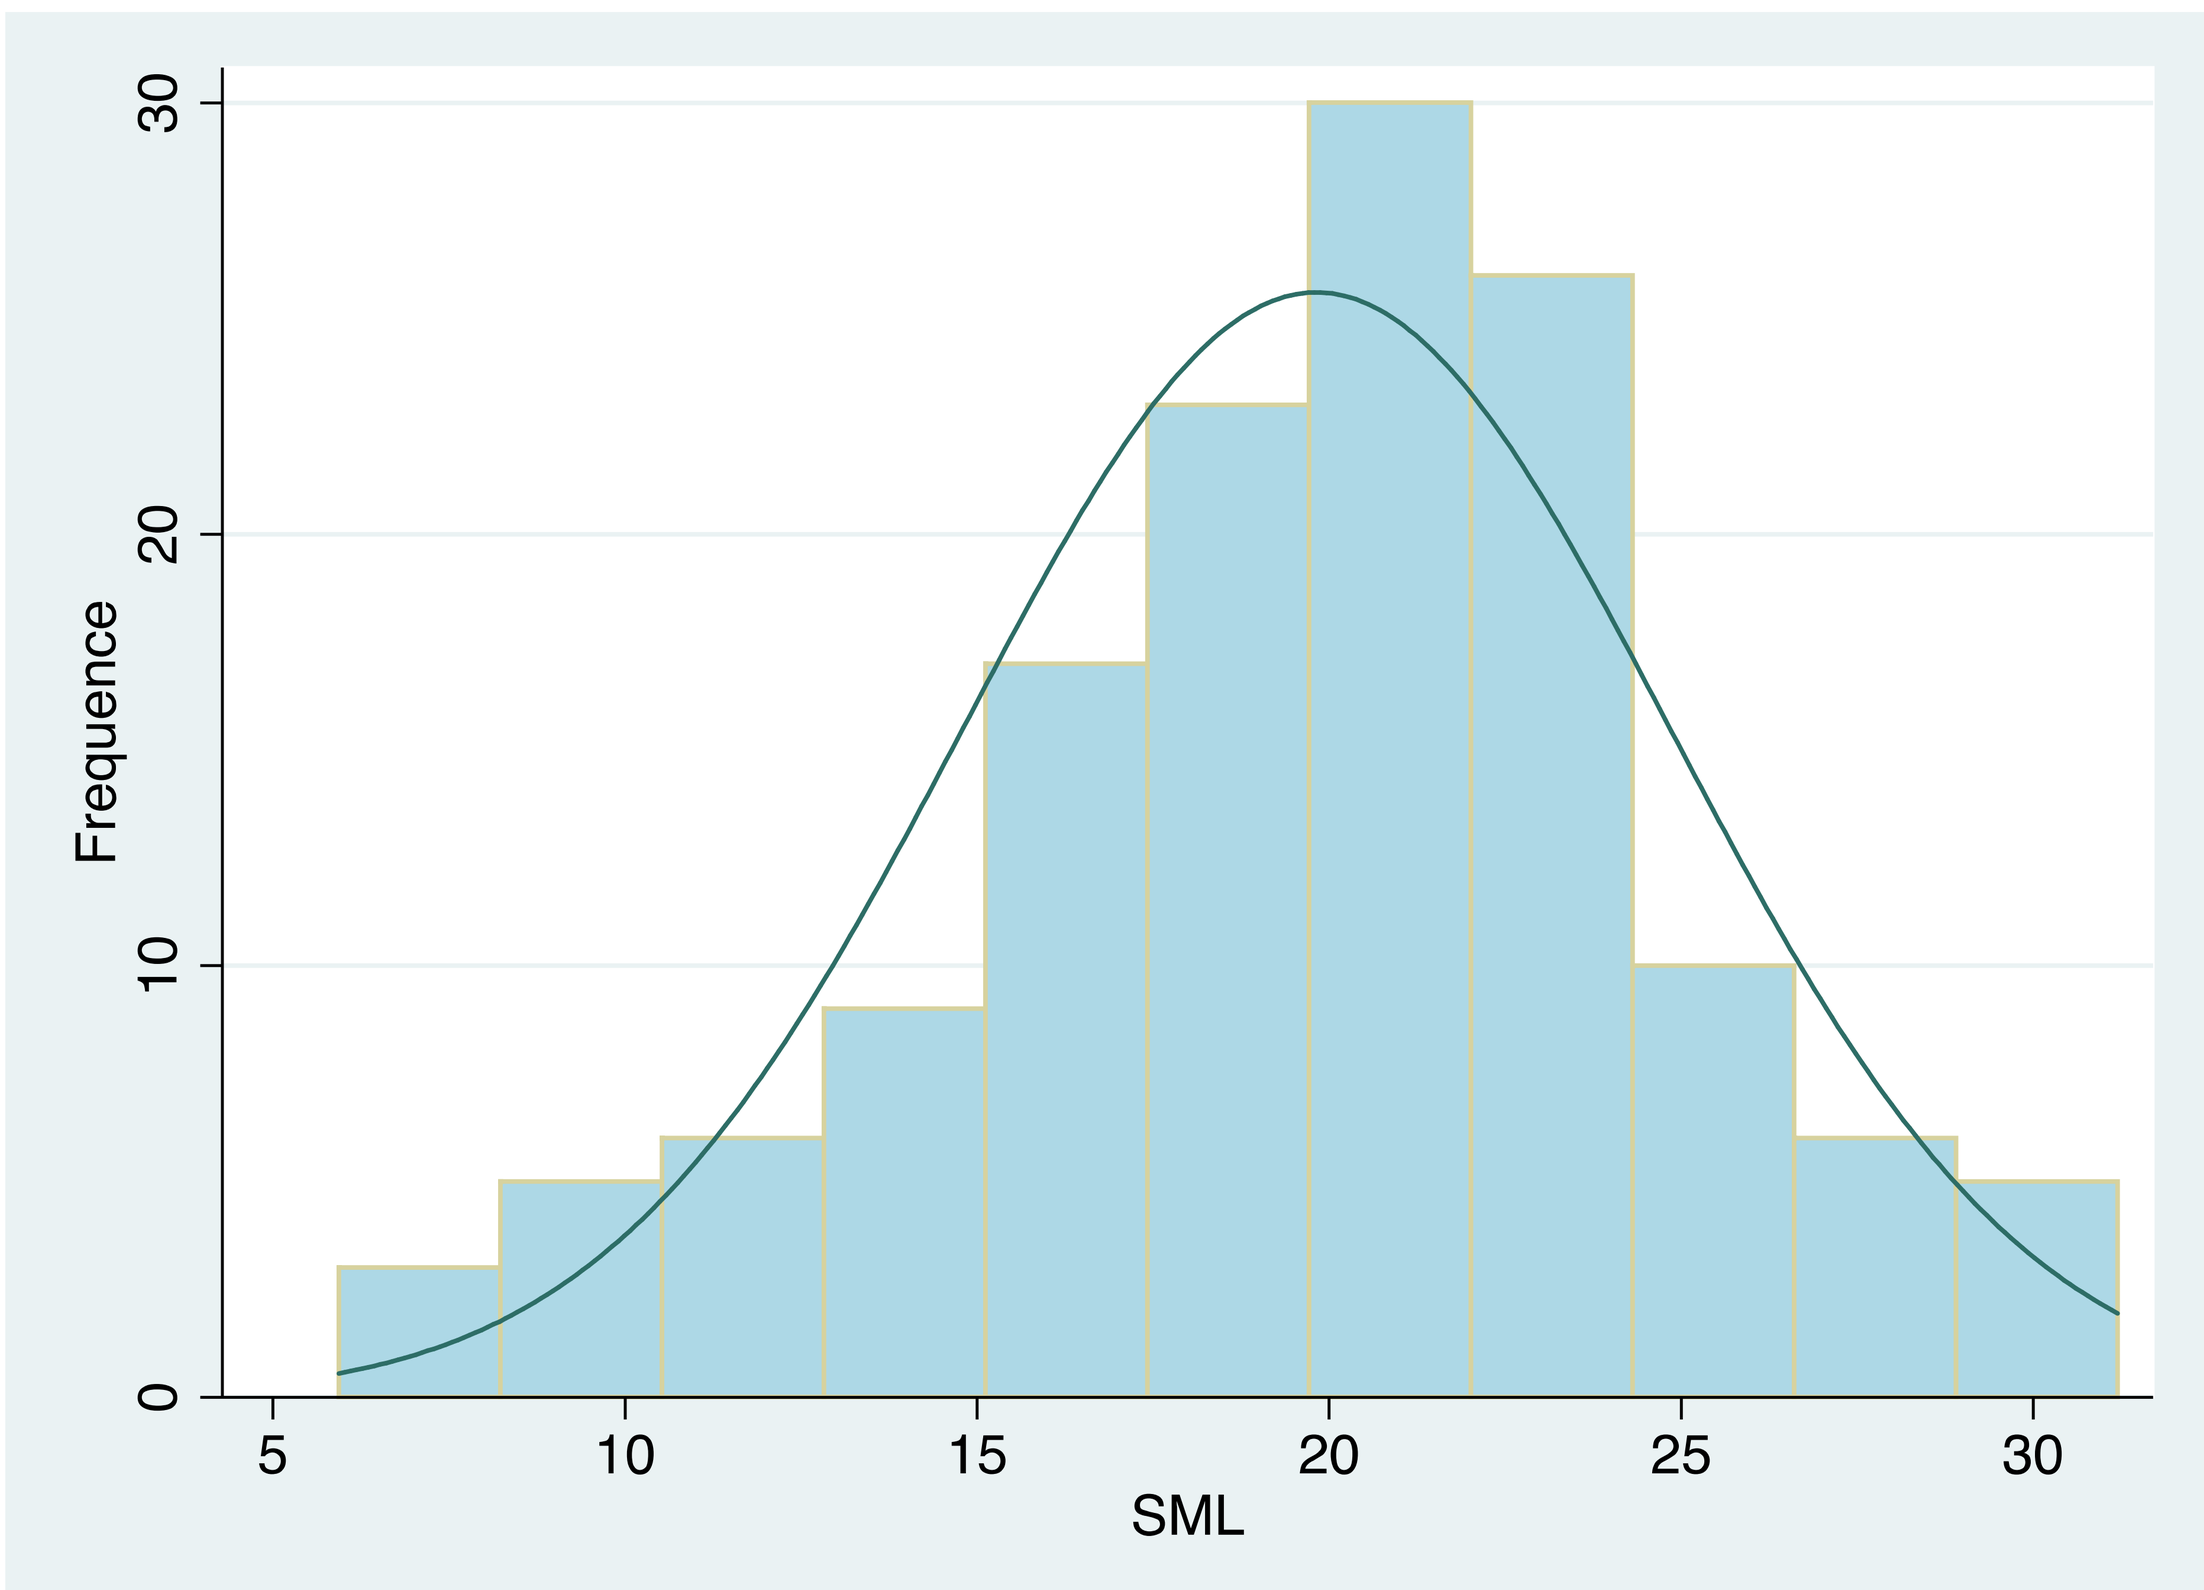
Frequence
SML

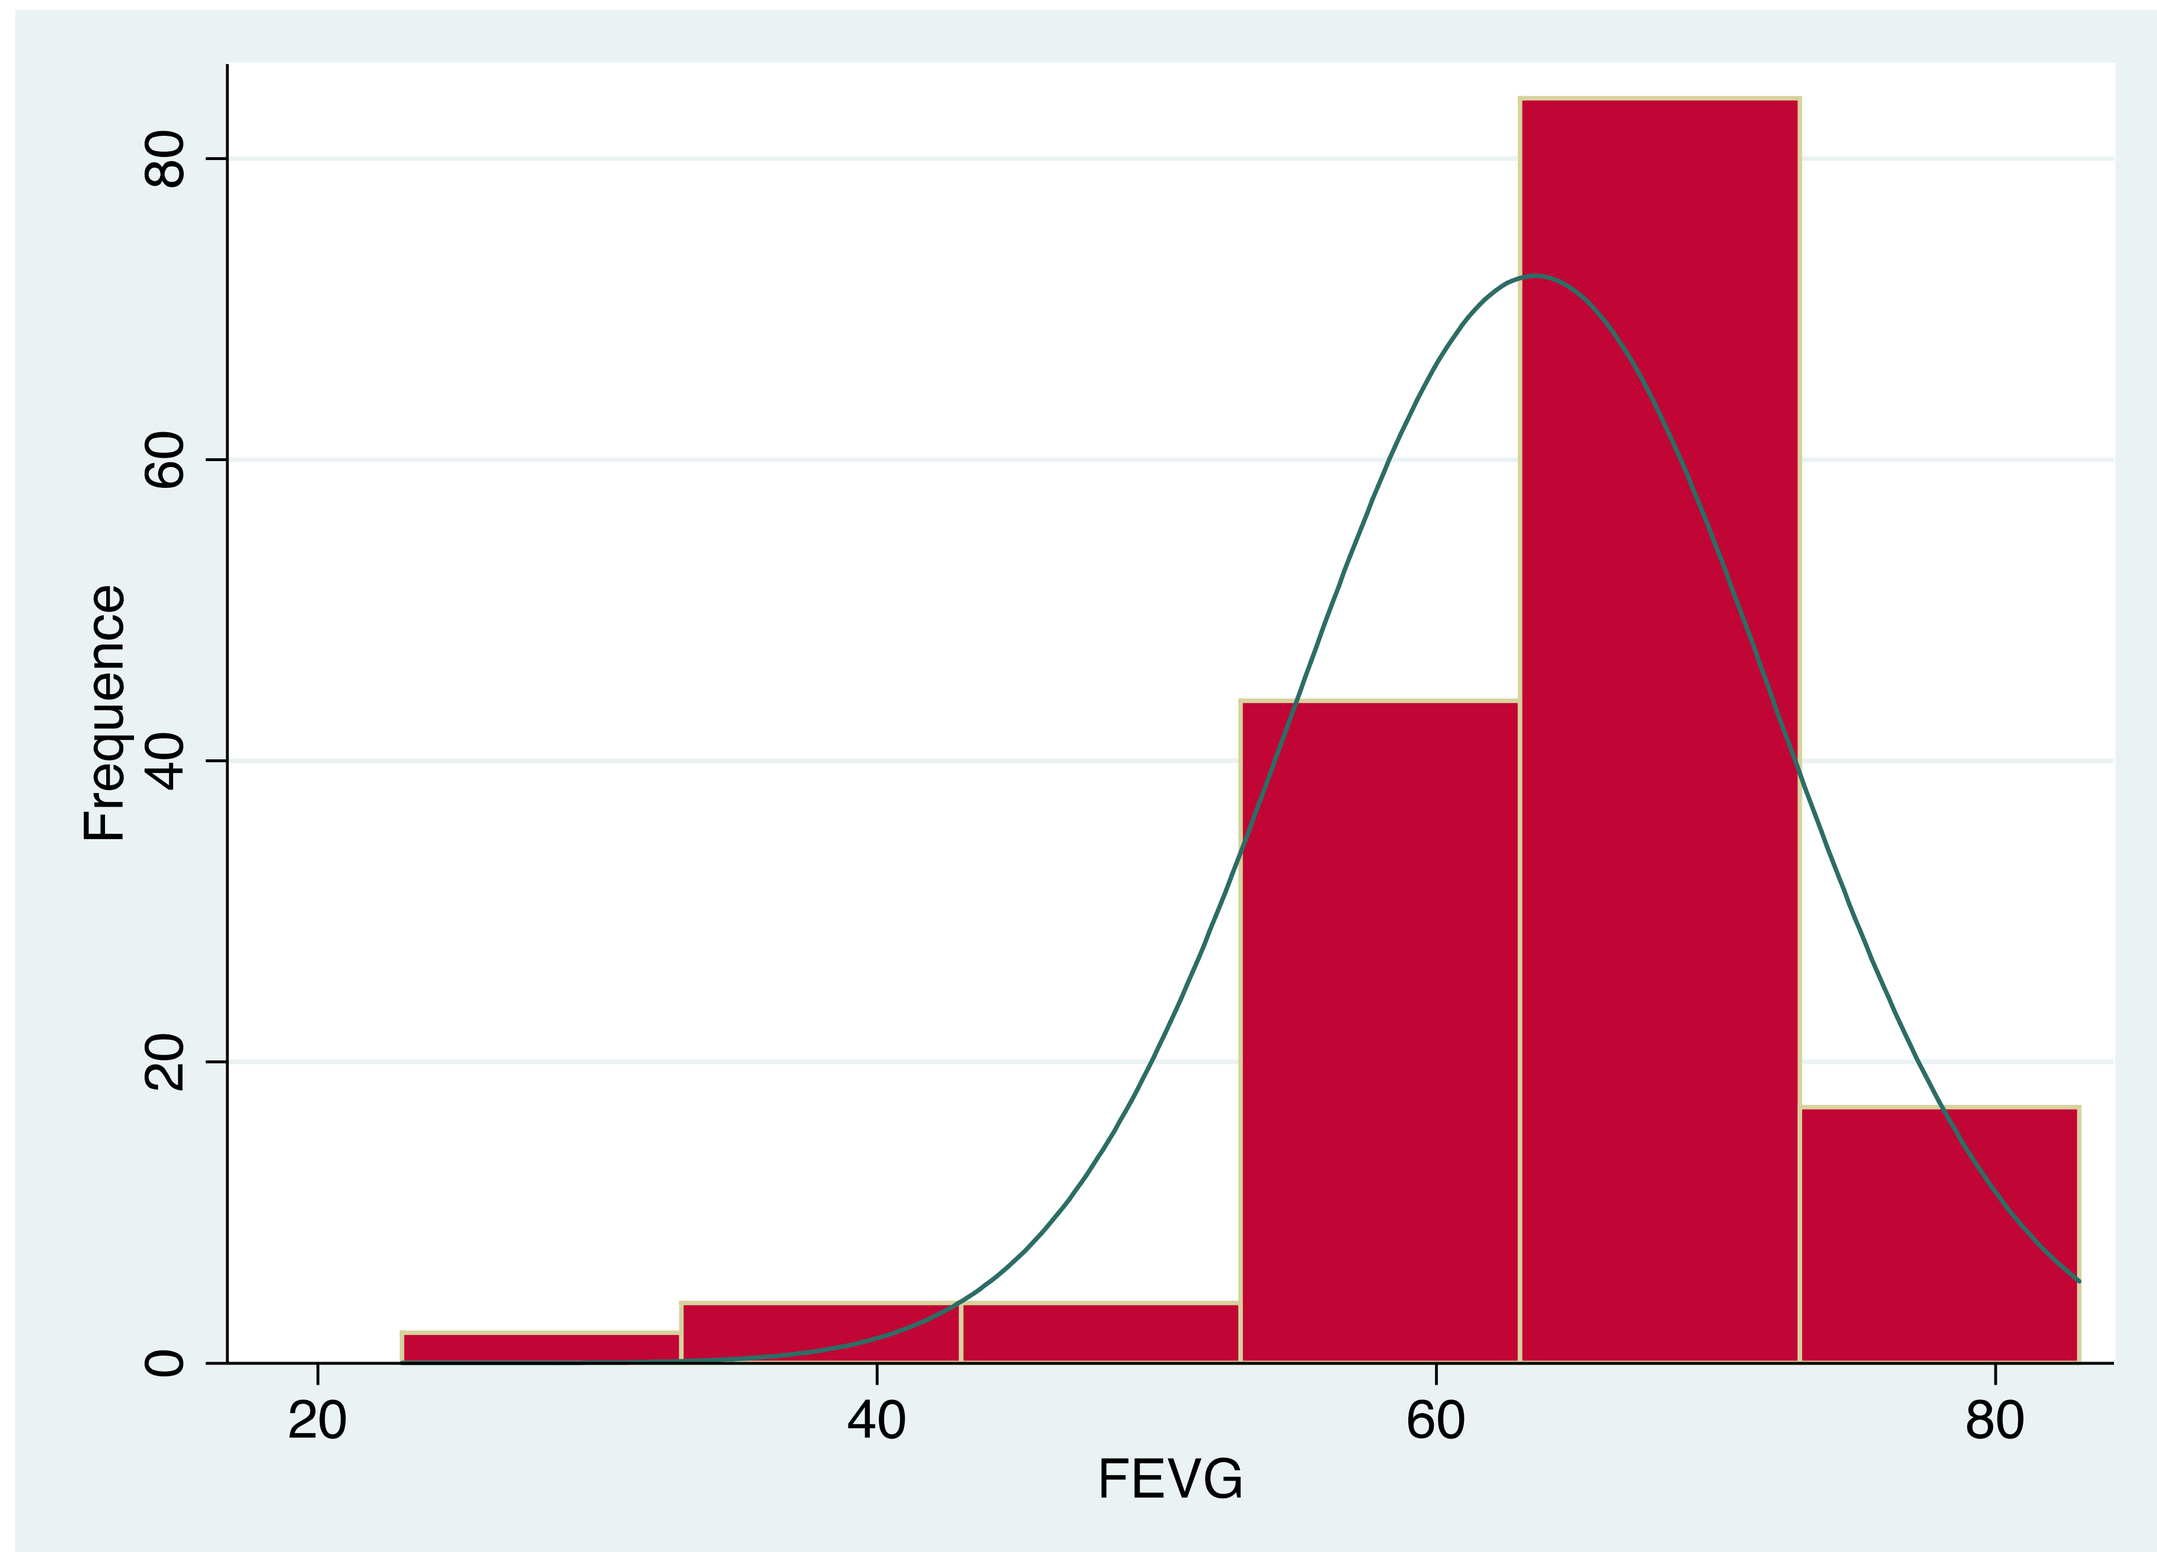
Frequence
FEVG

MATERIEL and METHODS

- **156 patients** included from 2003 to november 2017: all patients had genetically confirmed FRDA , referred to cardiology by neurogenetics department. Prospective follow up ended in may 2018
- Cardiac evaluation: clinical examination, 12 leads ECG and echocardiographic parameters included retrospective measures of GLS (4 chambers, TOMTEC Soft ware)
- **Primary Endpoint: a**ll causes death
- **Composite endpoint**: death + cardiovascular events (stroke, atrial fibrillation and heart failure)
- Cox analysis: Predictive factors of mortality and cardiovascular events
- Youden index: Cut off value of strain to diagnose FRDA cardiopathy

RESULTS (2): EVENTS

a) EVENTS, Follow up : 7.7 ± 4.0 y

Death *	N= 17 (11%)
Cardiovascular death	N=13 (8%)
Atrial Fibrillation *	N=13 (8%)
Stroke*	N=3 (2%)
Heart Failure*	N=7 (4%)
Composite Endpoint	N = 28 (18%)

b) Factors Associated with All-cause Mortality

	Univariate Cox Model			Multivariate Cox model		
Parameters	N	Hazard-ratio (95% CI)	P-value	N	Hazard-ratio (95% CI)	P-value
GLS (%)	141	1.20 (1.10;1.32)	0.0001*	139	1.05 (0.94;1.18)	0.4031
Age (years)	156	0.98 (0.93;1.03)	0.3752	139	0.99 (0.92;1.97)	0.7975
Age at diagnostic (years)	155	0.84 (0.76;0.94)	0.0023*	139	0.94 (0.83;1.06)	0.3234
GAA on the shorter allele	156	1.28 (1.11;1.47)	0.0008*	139	1.17 (0.98;1.40)	0.0795
LVMI (g/m²)	156	1.02 (1.01;1.04)	0.0078*	139	1.01 (0.99;1.03)	0.3131
LVEF (%)	155	0.88 (0.85;0.92)	<.0001*	139	0.93 (0.88;0.99)	**0.0213***

c) Factors associated with composite endpoint

	Univariate Cox model*			Multivariate Cox Model†		
Parameters	N	Hazard-ratio (95% CI)	P-value	N	Hazard-ratio (95% CI)	P-value
GLS (%)	139	1.16 (1.07;1.25)	0.0002	137	1.02 (0.93;1.12)	0.6258
Age (years)	154	0.98 (0.95;1.02)	0.3935	137	1.03 (0.97;1.08)	0.3679
Age at diagnostic (years)	153	0.87 (0.80;0.94)	0.0008	137	0.90 (0.82;1.00)	**0.0427***
Gaa on the shorter allele	154	1.18 (1.07;1.30)	0.0011	137	1.09 (0.96;1.23)	0.1659
LVMI (g/m²)	154	1.02 (1.01;1.03)	0.0003	137	1.02 (1.00;1.03)	0.0667
LVEF (%)	153	0.90 (0.87;0.93)	<.0001	137	0.94 (0.90;0.99)	**0.0093***

CONCLUSION

- **GLS is a predictor of morbimotality in FRDA**
- **GLS is not superior to LVEF as a prognostic marker**
- **Proposed cut off value of GLS to diagnose FRDA cardiopathy: -15,4%**
- **Prospective studies are mandatory to asses the early predictive value of GLS in FRDA**

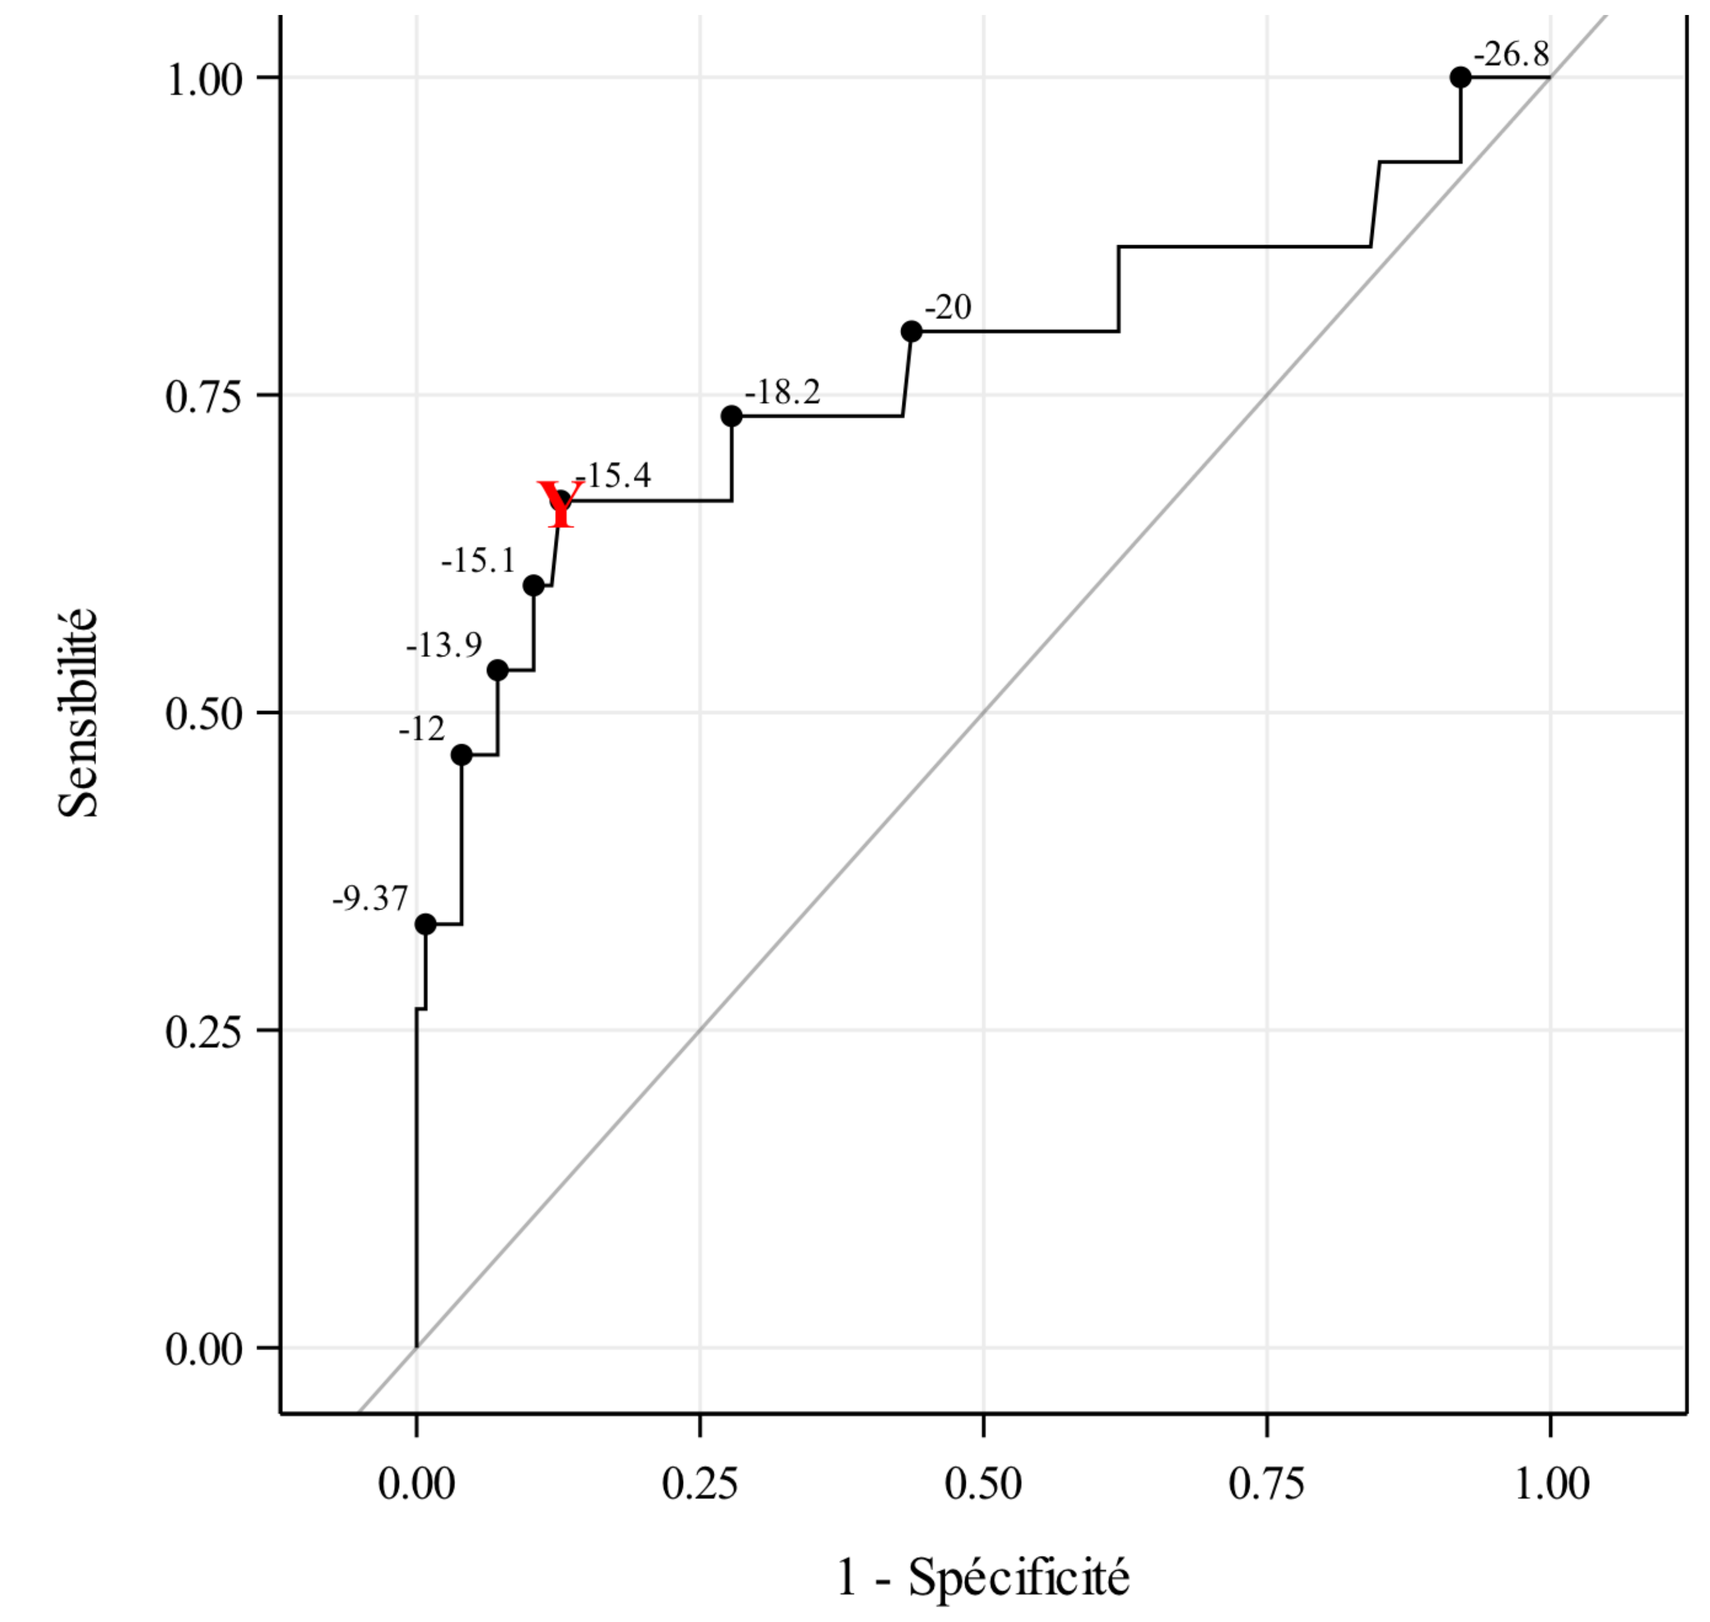
Sensibilité
1 - Spécificité
-9.37
-12
-13.9
-15.1
-15.4
-18.2
-20
-26.8

The authors have no conflicts of interest